QL535, A Novel CD2-Costimulating T Cell Engager Targeting PSMA-Positive Prostate Cancer Matching CD28 Trispecific Antibody in Cytotoxicity, Minimizing Cytokine Release and T-Cell Exhaustion

Xiao Liu[1], Shenda Gu[1], Ke Liu[2], Joshua Morrissette[1], Jiantao Kong[2], Guangjie Guo[1], Yuxia Wang[2], Xin Zhang[2], Jiaming Cao[2], Moyan Hu[2], Min Sun[2], Ruixue Ma[2], Jane Chen[1], Nathan Ngo[1], Hejie Zhu[2], Liwei Wang[2], Yan Fang[2], Hao Liu[2], Shuyong Zhao[2], Shihao Chen[1]

QLSF Biotherapeutics, Inc., South San Francisco, CA, USA | 2. QILU Pharmaceutical Co., Ltd., Jinan, China | 3. QILU Pharmaceutical Co., Ltd., Shanghai, China

Abstract 3495
AACR 2025

Introduction

Prostate cancer is a leading cause of cancer-related mortality in men, highlighting the need for new therapeutic options due to resistance to current treatments[1,2]. PSMA (Prostate-Specific Membrane Antigen) is overexpressed in prostate cancer tissues compared to normal prostatic tissue rendering it an ideal target for prostate cancer therapies, enabling precise delivery of therapeutic agents to cancer cells[3].

We developed QL535, a novel trispecific antibody in a 2+1+1 format designed to bridge tumor cells via PSMA and activate T cells through CD3 (signal 1) and CD2 (signal 2) via its ligand CD58. This design aims to stabilize the immune synapse and enhance T-cell cytotoxicity. Additionally, it overcomes T-cell exhaustion, with fine-tuned CD3 affinity to improve safety and reduce the risk of cytokine release syndrome (CRS).

Our TECOS 'T Cell Engager with Co-Stimulation' Platform with CD2-Costimulating

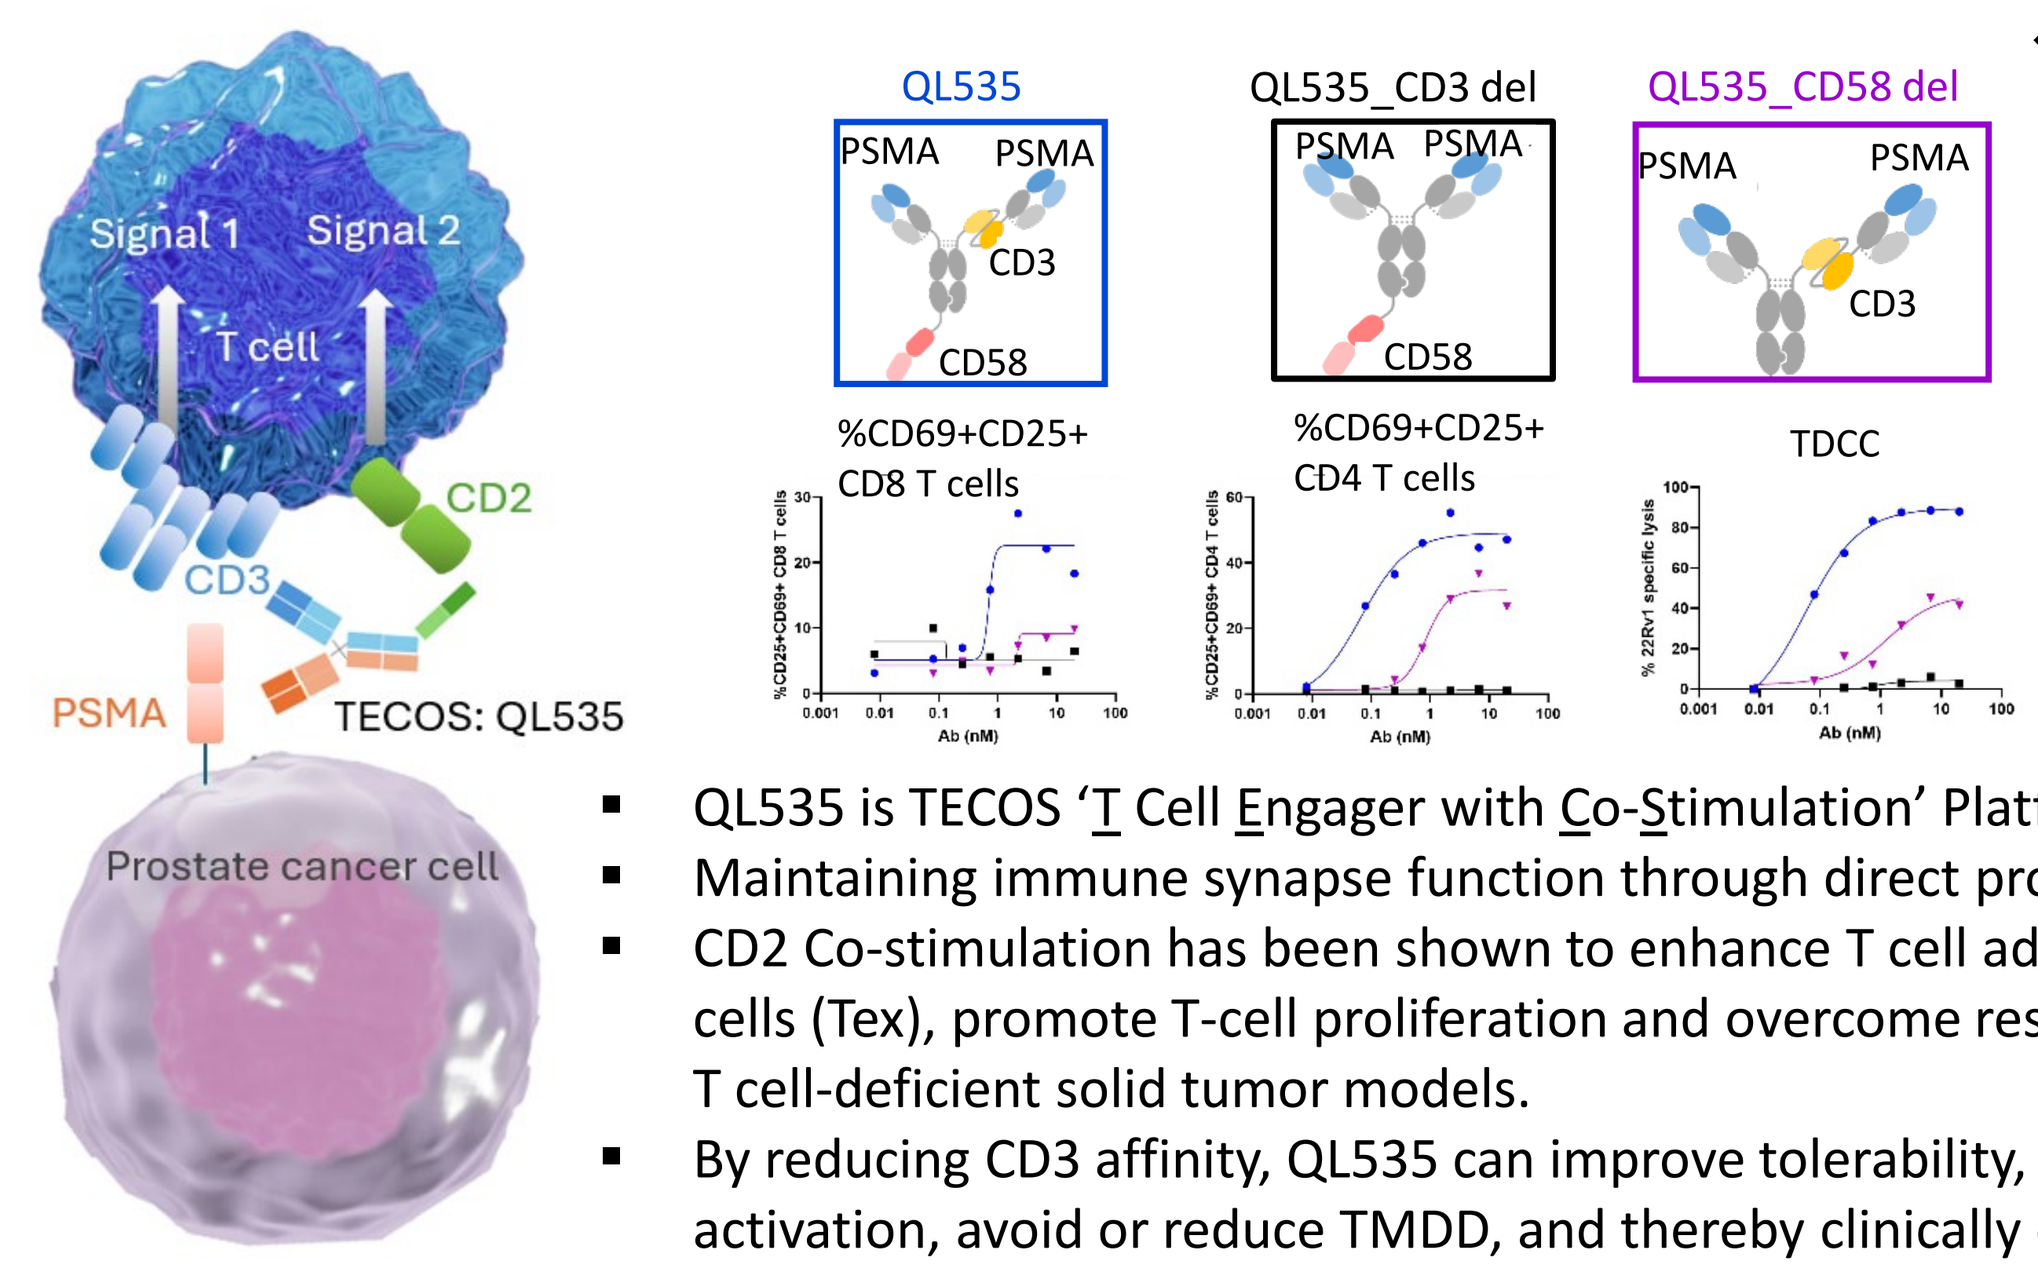
Signal 1
Signal 2
T cell
CD2
CD3
PSMA
TECOS: QL535
Prostate cancer cell
QL535
QL535_CD3 del
QL535_CD58 del
PSMA
CD3
CD58
%CD69+CD25+ CD8 T cells
%CD69+CD25+ CD4 T cells
TDCC

❖ Comparative analysis was conducted with three trispecific antibody formats targeting CD28 and three formats targeting CD2, resulting in a lead CD2 trispecific antibody as QL535, alongside a CD28 trispecific antibody as a control.

- QL535 is TECOS 'T Cell Engager with Co-Stimulation' Platform with CD2-Costimulating.
- Maintaining immune synapse function through direct provision of CD2 co-stimulation.
- CD2 Co-stimulation has been shown to enhance T cell adaptability, restore exhausted T cells (Tex), promote T-cell proliferation and overcome resistance to T cell therapy (TCE) in T cell-deficient solid tumor models.
- By reducing CD3 affinity, QL535 can improve tolerability, minimize antigen non-specific activation, avoid or reduce TMDD, and thereby clinically control safety.

Target Discovery and Evaluation

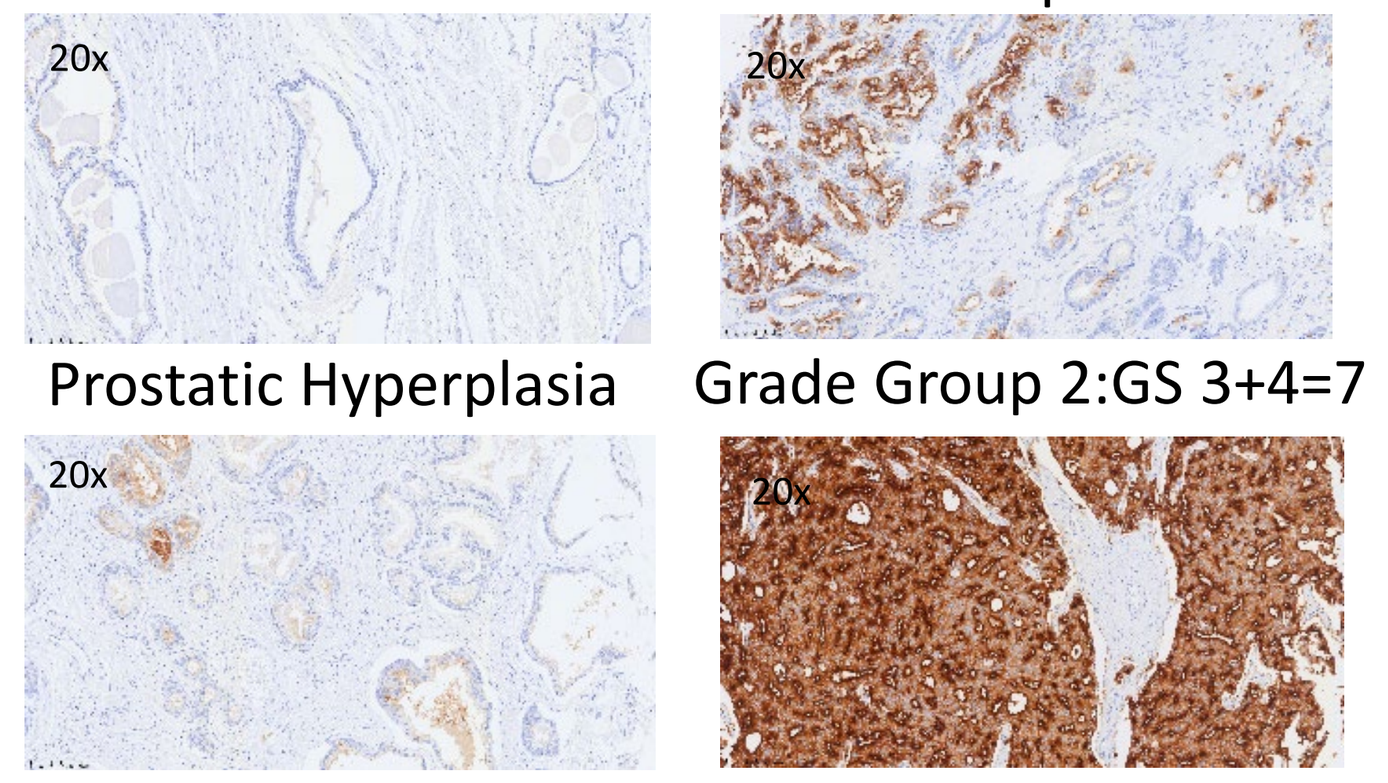
Normal Prostatic Tissue
Grade Group 1:GS 3+2=5
Prostatic Hyperplasia
Grade Group 2:GS 3+4=7
20x

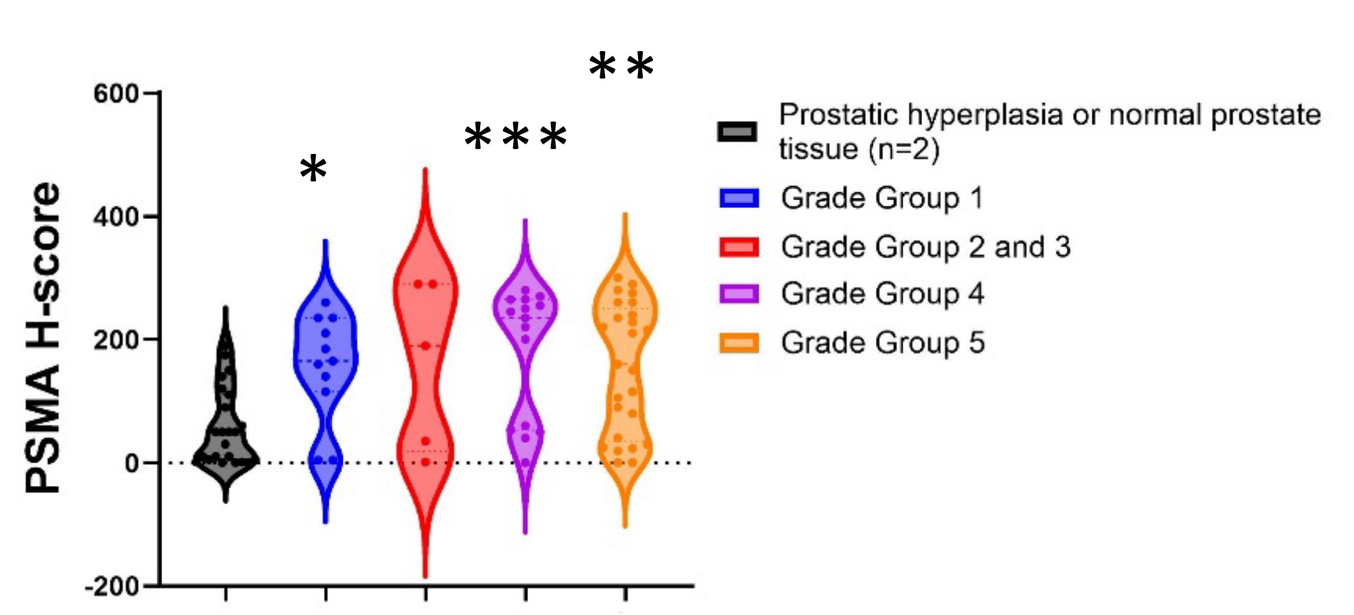
PSMA expression (IHC H-score)
PSMA H-score
Prostatic hyperplasia or normal prostate tissue (n=2)
Grade Group 1
Grade Group 2 and 3
Grade Group 4
Grade Group 5

We evaluated PSMA expression in 77 Chinese prostate cancer patients using immunohistochemistry (IHC), confirming its expression in Chinese malignant prostate tissues.

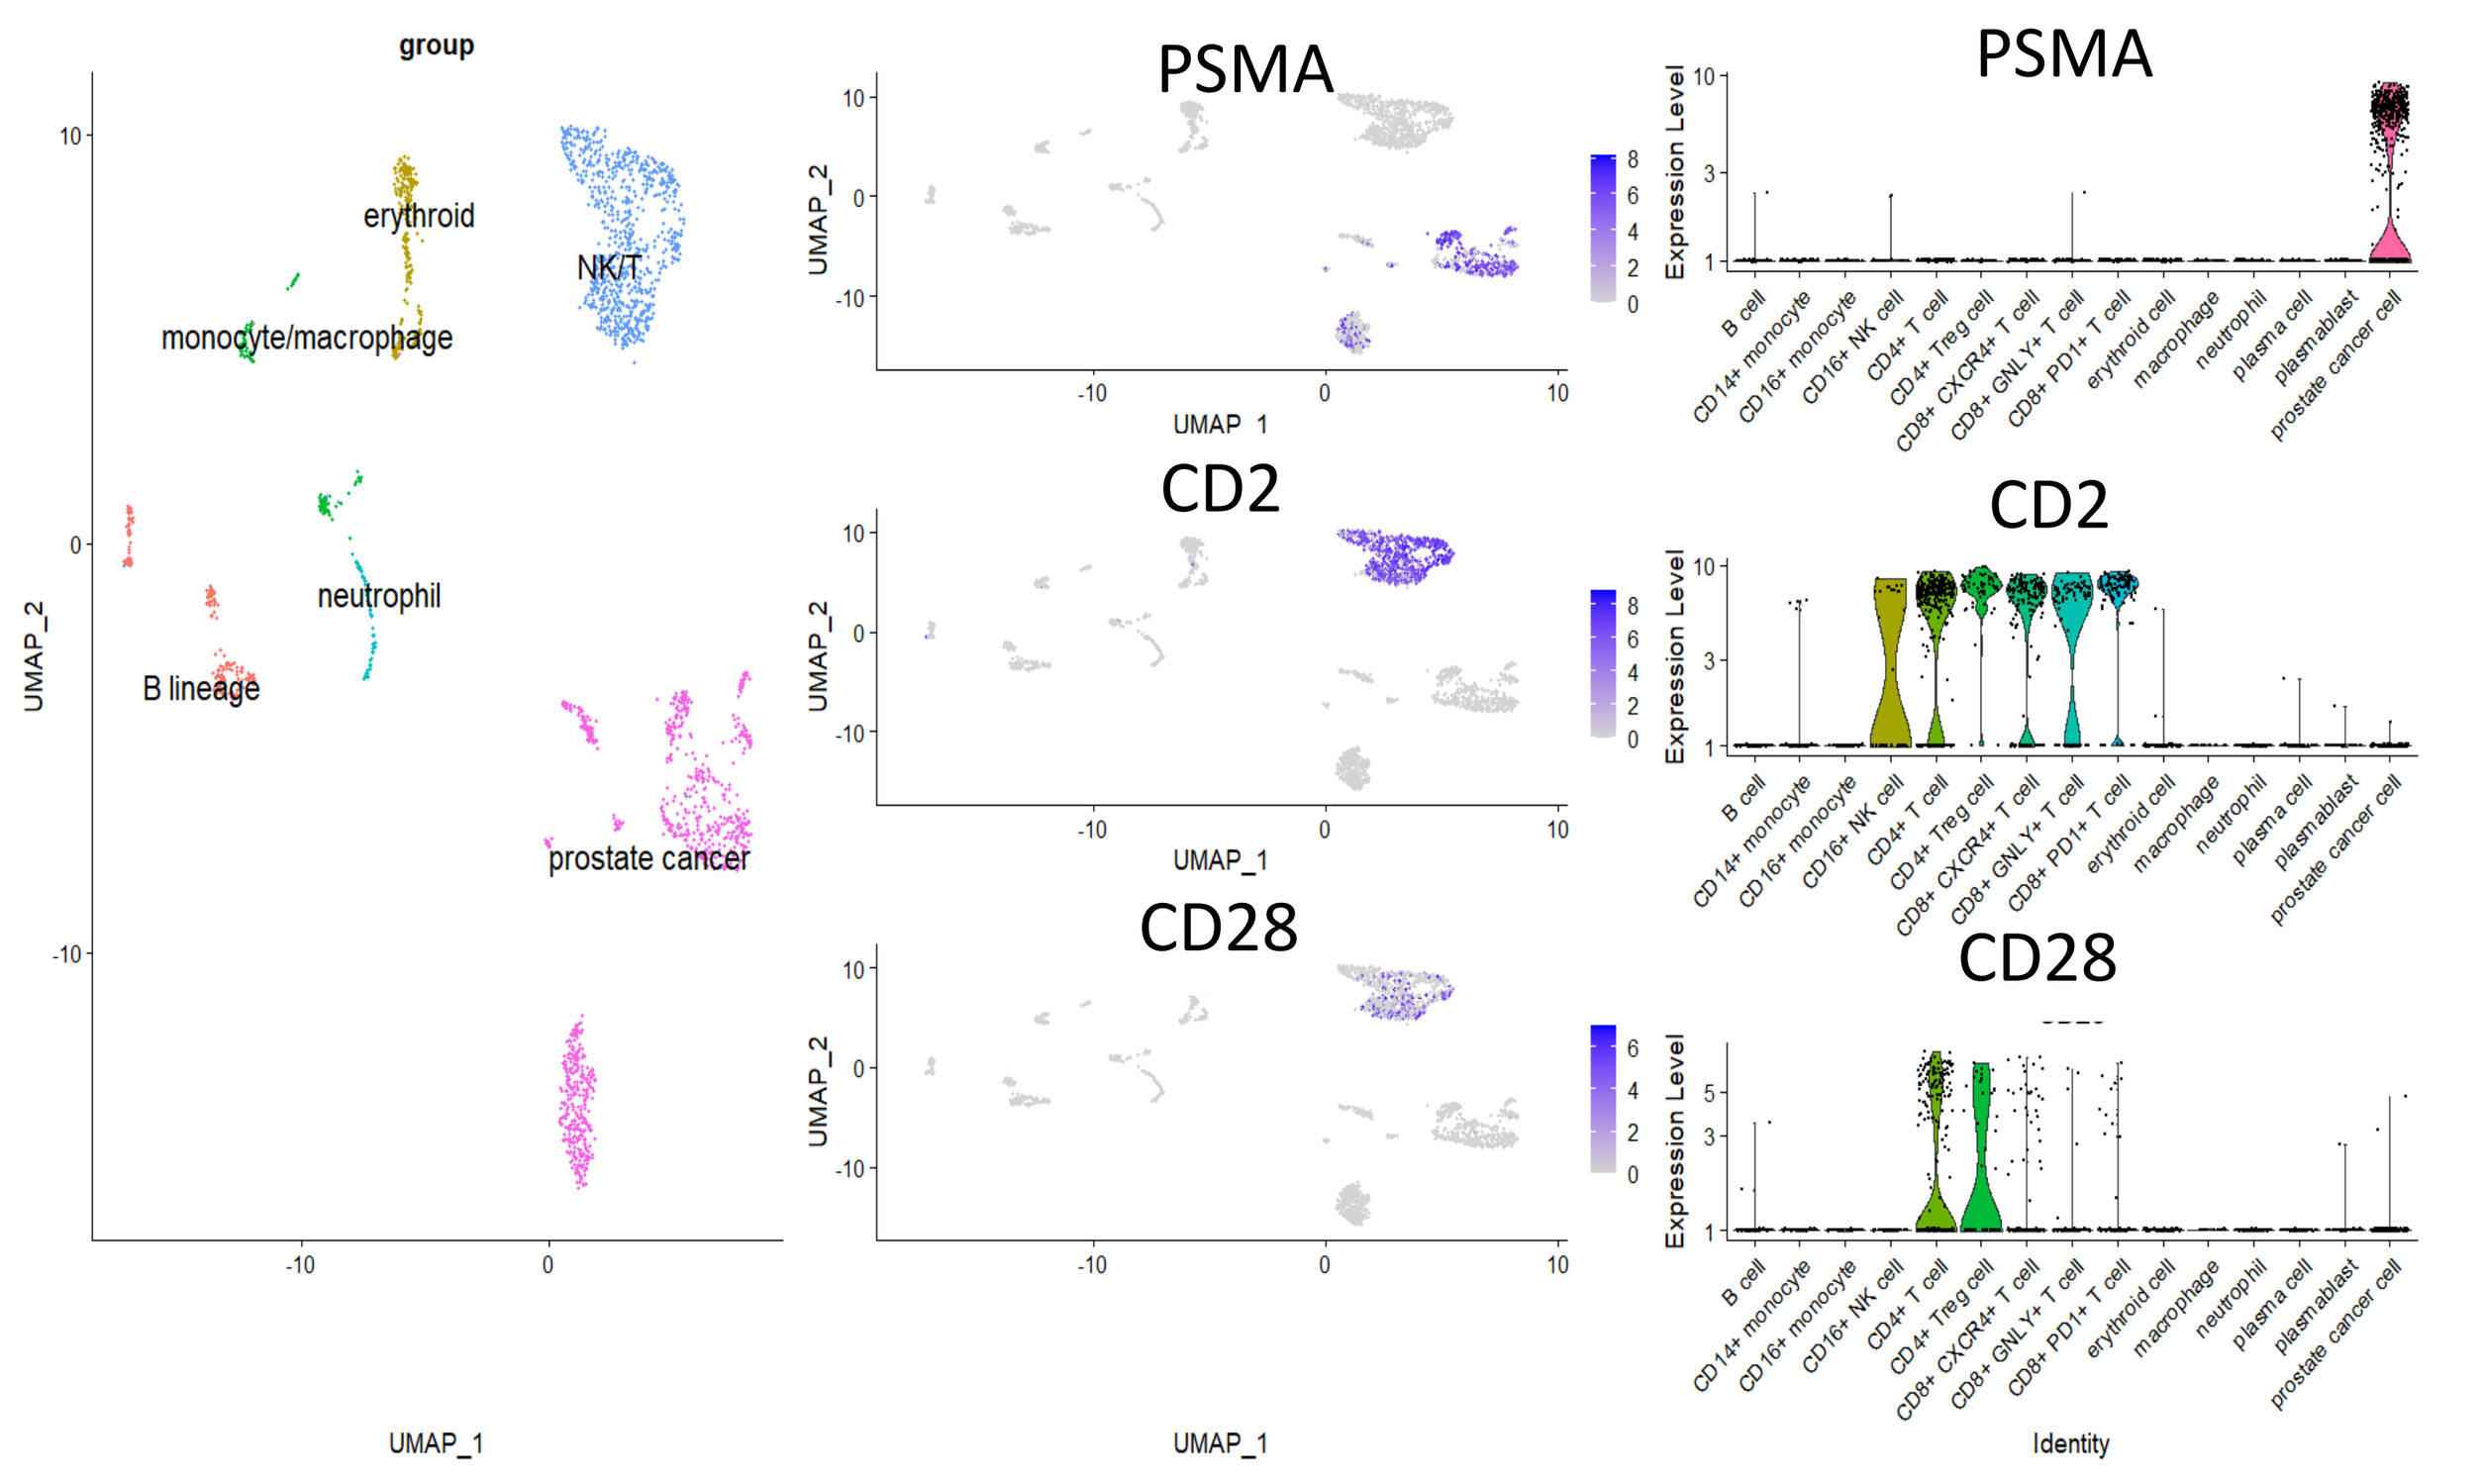
group
erythroid
NK/T
monocyte/macrophage
neutrophil
B lineage
prostate cancer
PSMA
CD2
CD28
UMAP_1
UMAP_2
Expression Level
Identity

Analysis of single-cell RNA sequencing data from 2,170 cells across 14 mCRPC patients revealed that CD2 expression on tumor-infiltrating lymphocytes (TILs), particularly CD8⁺ T cells, exceeds that of CD28, underscoring the potential for CD2 targeting in enhancing antitumor immunity.

QL535 Exhibits Favorable Drug Developability

1. QL535 exhibits similar CD2 and CD3 binding affinity to both human and cynomolgus by SPR analysis.

CD3_SPR

Specimen	ka (1/Ms)	kd (1/s)	K_D (M)
Recombinant Human CD3	3.36E+05	0.008552	2.54E-08
Recombinant Cynomolgus Monkey CD3	3.65E+05	0.008743	2.39E-08

CD2_SPR

Specimen	ka (1/Ms)	kd (1/s)	K_D (M)
Recombinant Human CD2	1.59E+05	0.004589	2.88E-08
Recombinant Cynomolgus Monkey CD2	1.52E+05	0.004454	2.93E-08

2. QL535 demonstrates strong stability in human serum.

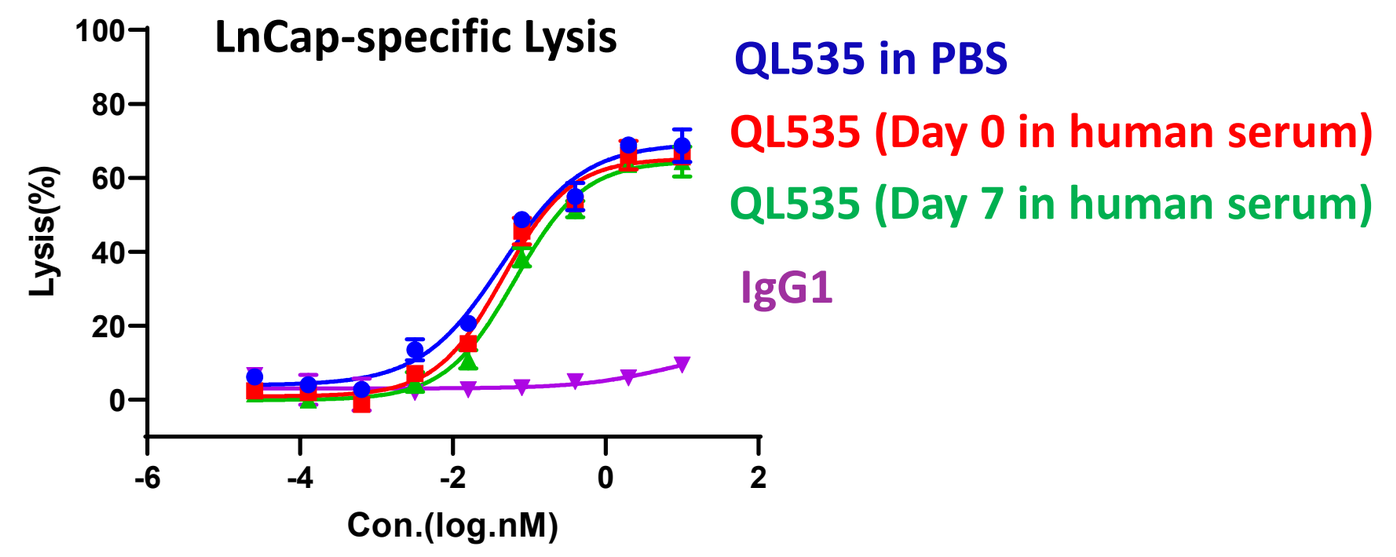
LnCap-specific Lysis
Lysis(%)
Con.(log.nM)
QL535 in PBS
QL535 (Day 0 in human serum)
QL535 (Day 7 in human serum)
IgG1

3. QL535 outperforms the other two formats in accelerated stability testing.

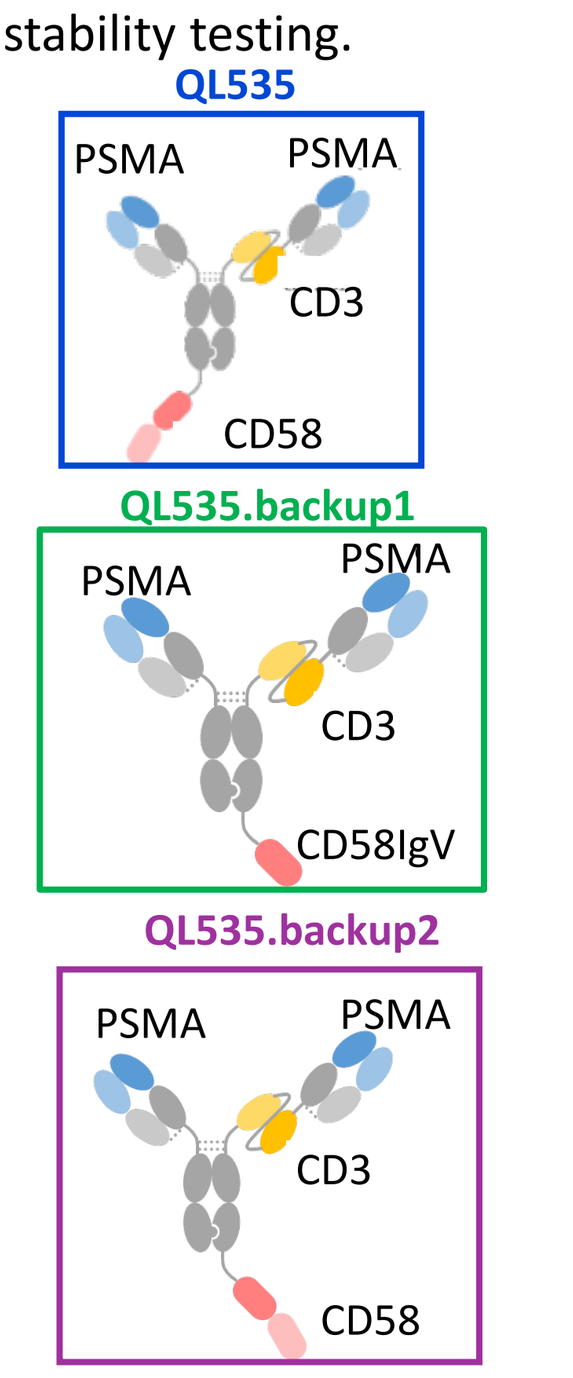
QL535
QL535.backup1
QL535.backup2
PSMA
CD3
CD58
CD58IgV

5mg/mL @ 42°C			
Week	%Agg	%Main	%Frag
0	1.2	98.4	0.4
1	2.4	96.4	1.2
2	3.7	94.3	2.0
3	4.5	92.9	2.6
4	5.4	91.4	3.2

5mg/mL @ 42°C			
Week	%Agg	%Main	%Frag
0	1.6	97.8	0.6
2	5.9	92.6	1.5
4	12.5	84.2	3.3

5mg/mL @ 42°C			
Week	%Agg	%Main	%Frag
0	1.6	97.7	0.7
1	2.6	95.3	2.1
2	2.8	94.6	2.6
3	3.8	92.6	3.6
4	6.0	90.5	3.5

QL535 Achieves CD28 Tri-specific Antibody Cytotoxicity with Reduced Cytokine Release

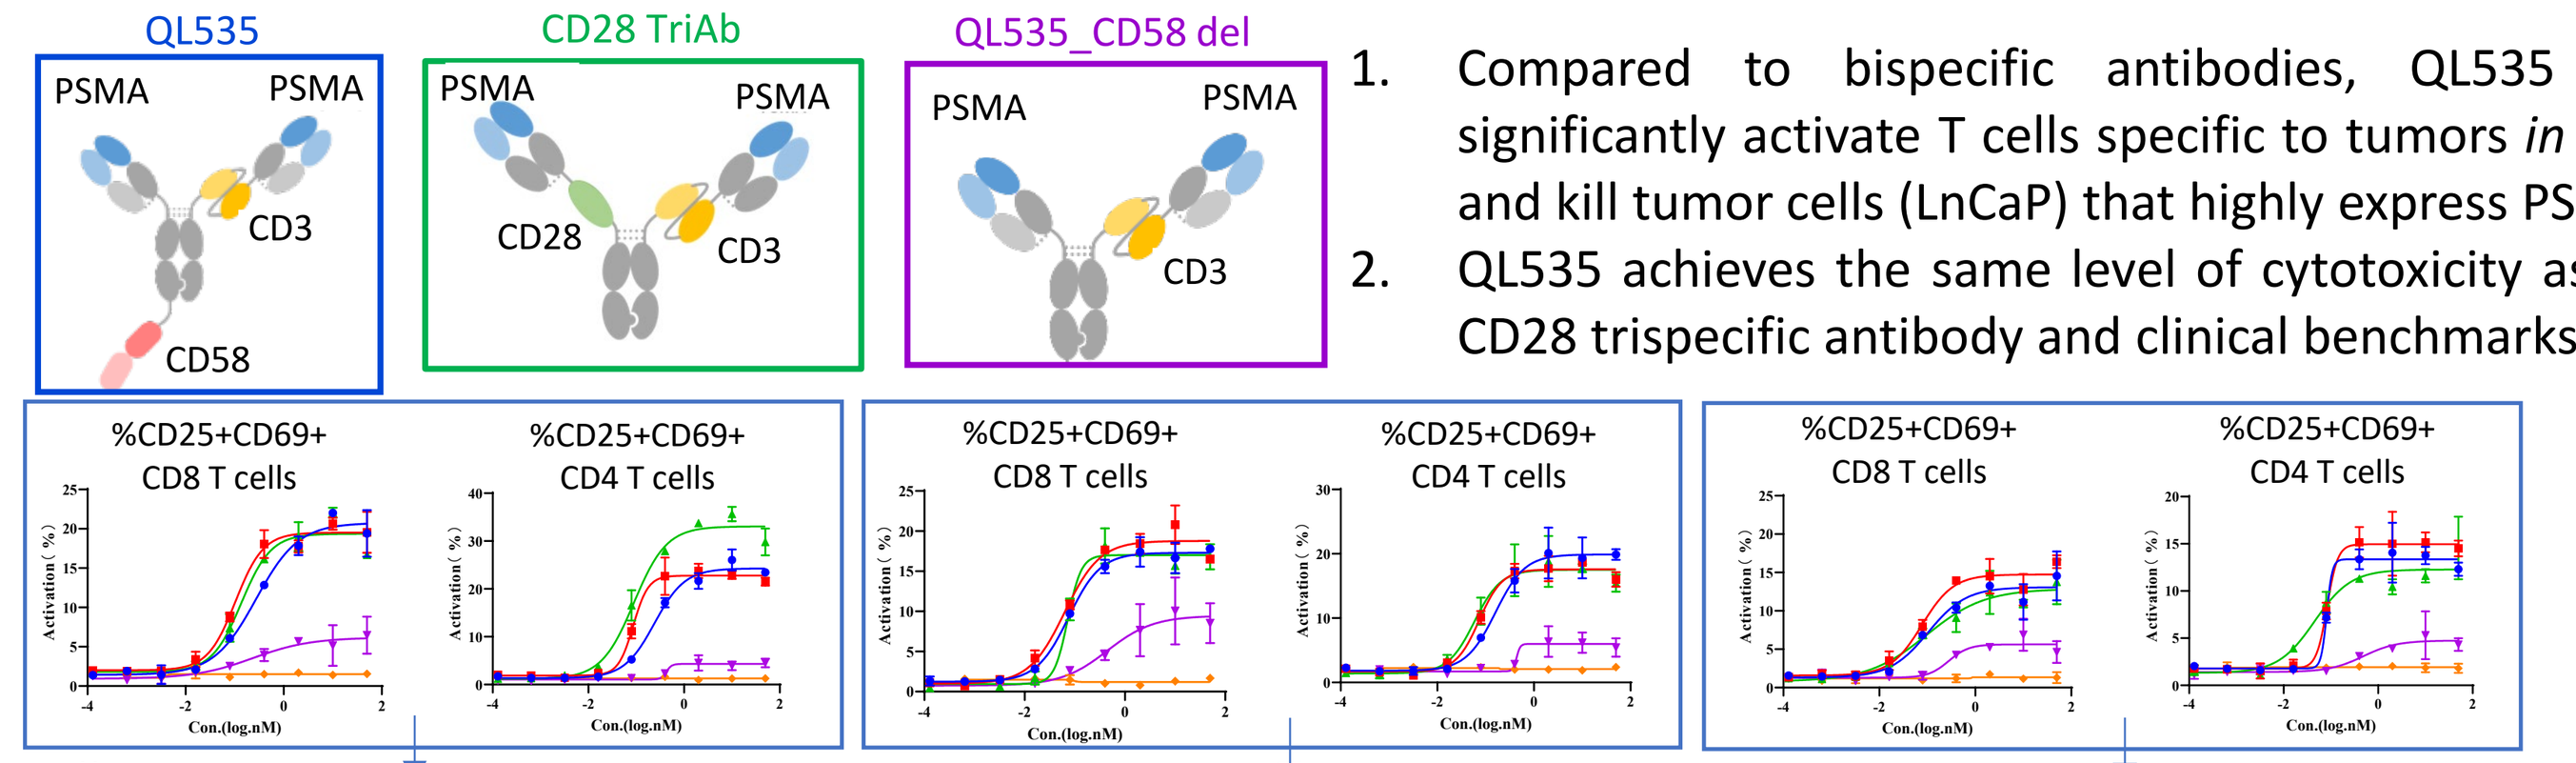
QL535
CD28 TriAb
QL535_CD58 del
PSMA
CD3
CD28
CD58
%CD25+CD69+ CD8 T cells
%CD25+CD69+ CD4 T cells

1. Compared to bispecific antibodies, QL535 can significantly activate T cells specific to tumors *in vitro* and kill tumor cells (LnCaP) that highly express PSMA.
2. QL535 achieves the same level of cytotoxicity as the CD28 trispecific antibody and clinical benchmarks

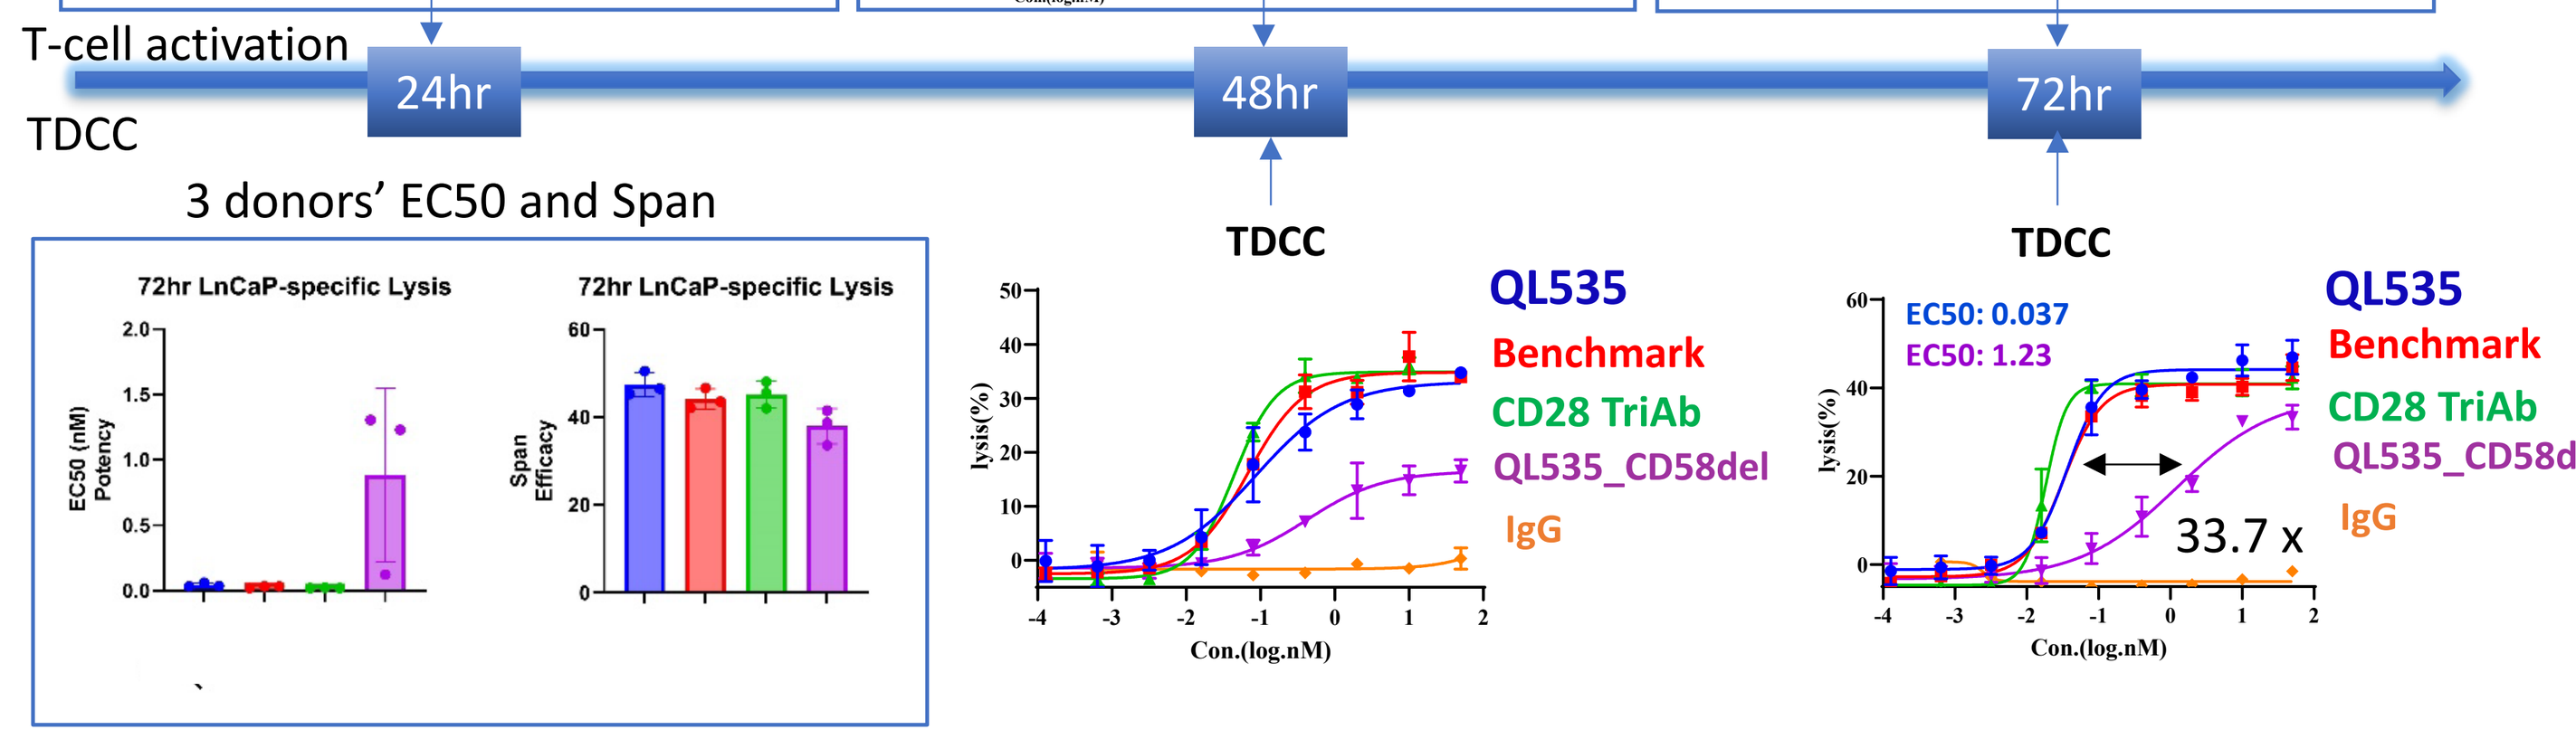
T-cell activation
24hr
48hr
72hr
TDCC
3 donors' EC50 and Span
72hr LnCaP-specific Lysis
EC50 (nM) Potency
Span Efficacy
QL535
Benchmark
CD28 TriAb
QL535_CD58del
IgG
EC50: 0.037
EC50: 1.23
33.7 x
lysis(%)
Con.(log.nM)

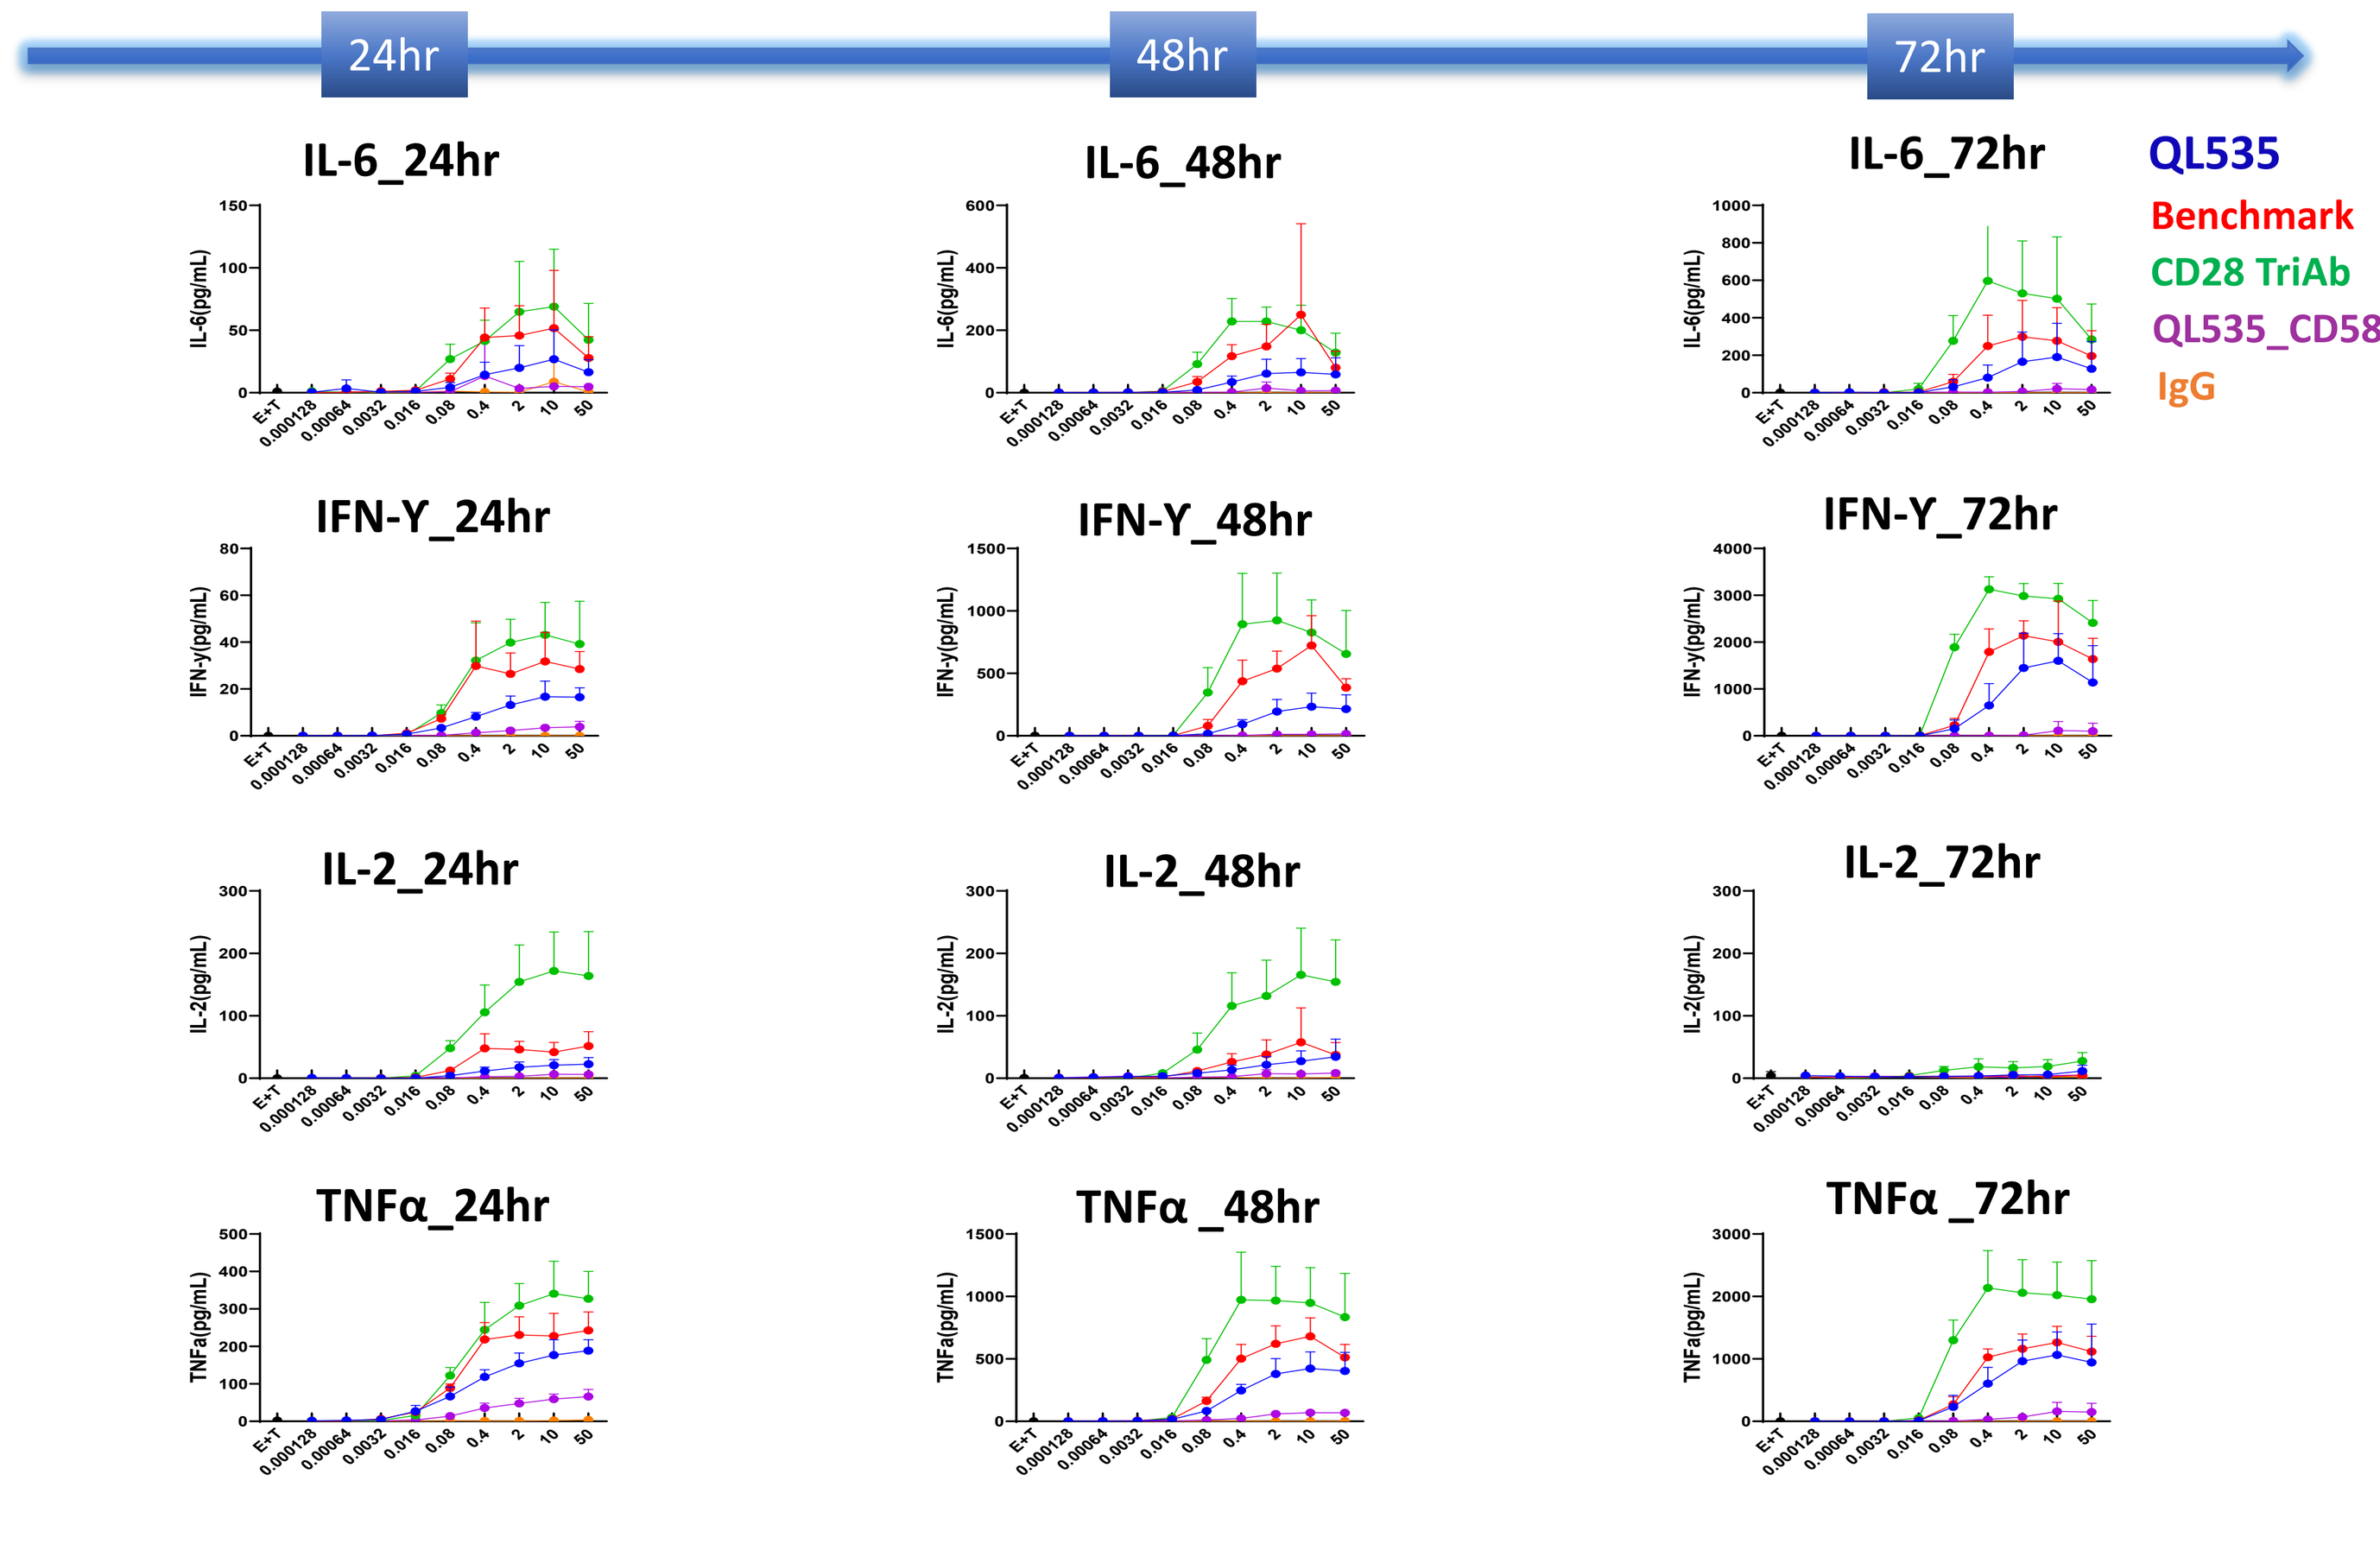
24hr
48hr
72hr
IL-6_24hr
IL-6_48hr
IL-6_72hr
IFN-Y_24hr
IFN-Y_48hr
IFN-Y_72hr
IL-2_24hr
IL-2_48hr
IL-2_72hr
TNFα_24hr
TNFα _48hr
TNFα _72hr
QL535
Benchmark
CD28 TriAb
QL535_CD58del
IgG

QL535's cytokine release levels (IL-6, IFN-y, IL-2, TNFa) are lower than those of clinical benchmark and CD28 trispecific antibody at the same level of cytotoxicity.

QL535 can Amplify the pPLC-γ1 Signaling Pathway to Facilitate T-Cell Activation

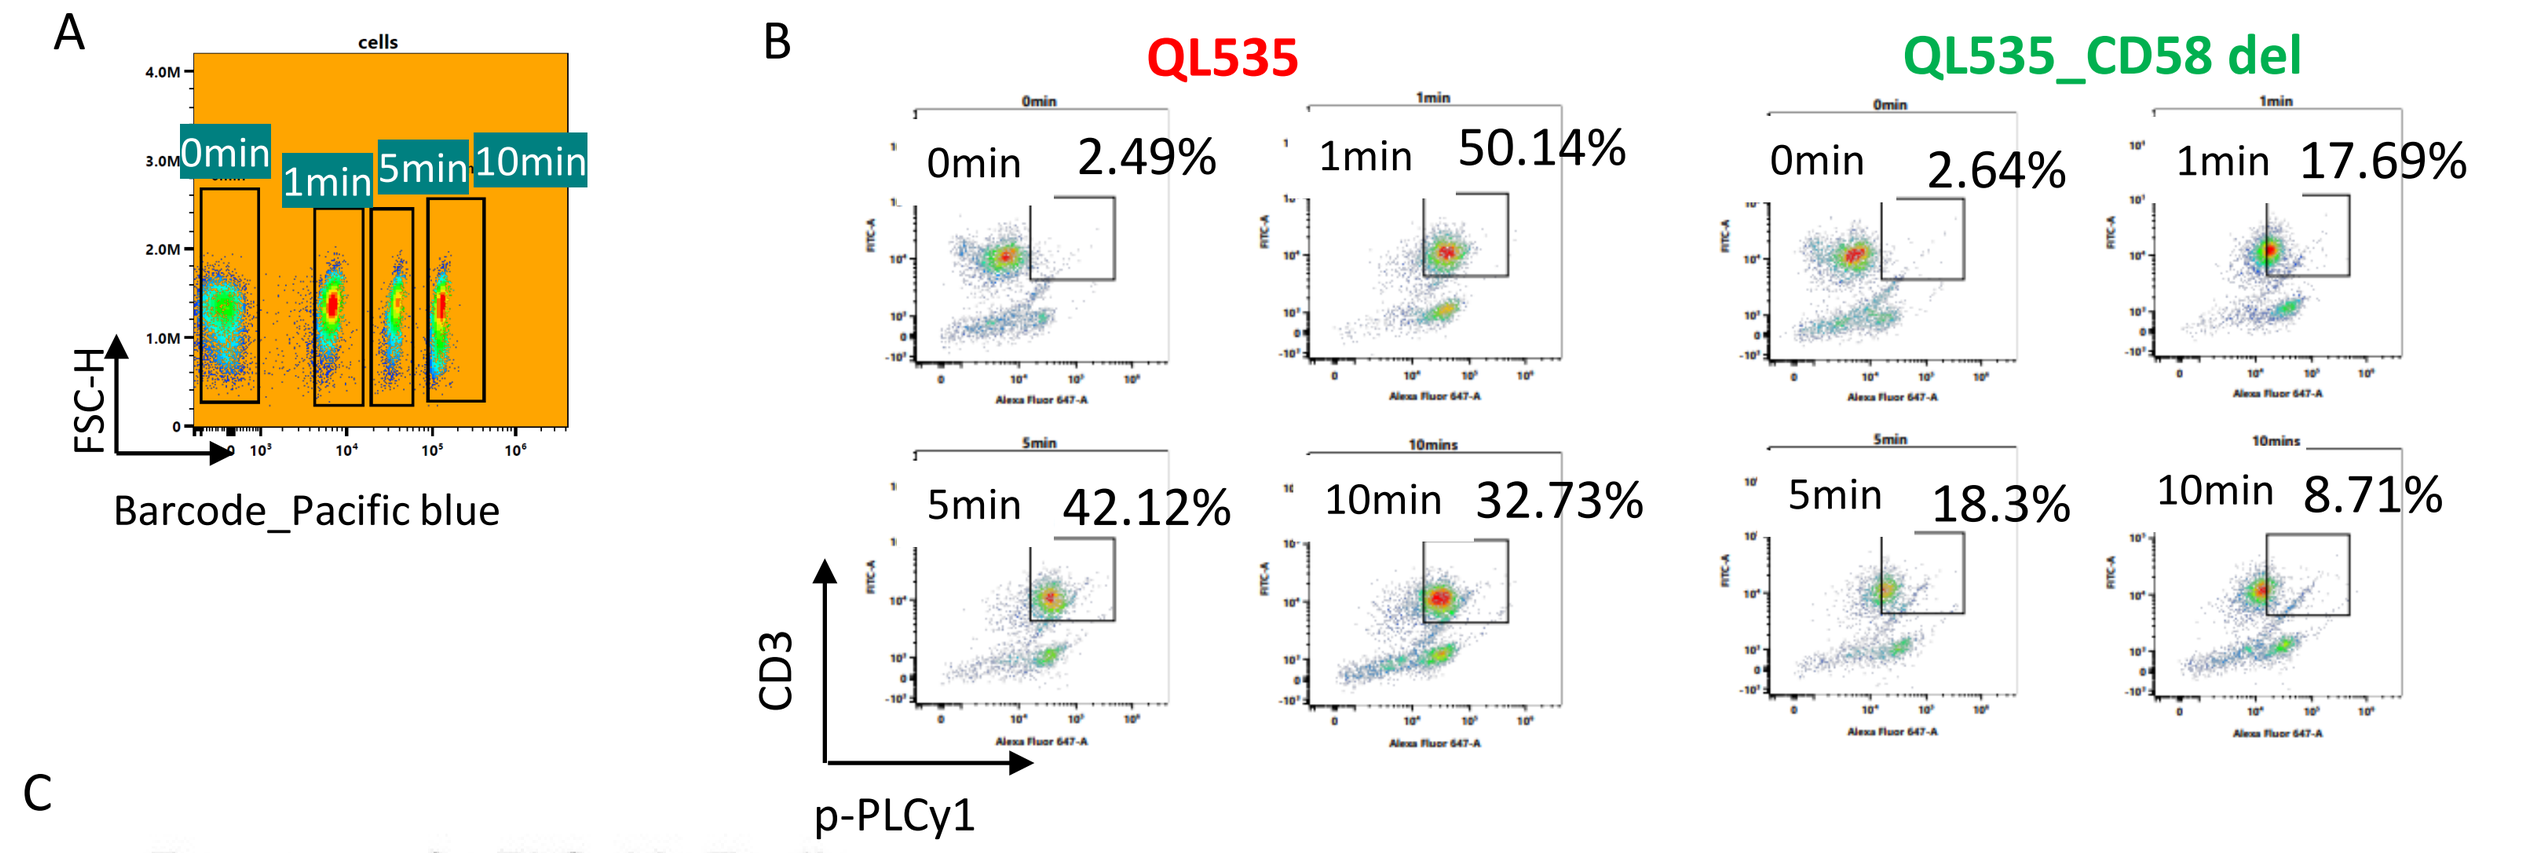
A
cells
0min
1min
5min
10min
FSC-H
Barcode_Pacific blue
B
QL535
QL535_CD58 del
0min 2.49%
1min 50.14%
5min 42.12%
10min 32.73%
0min 2.64%
1min 17.69%
5min 18.3%
10min 8.71%
CD3
p-PLCy1

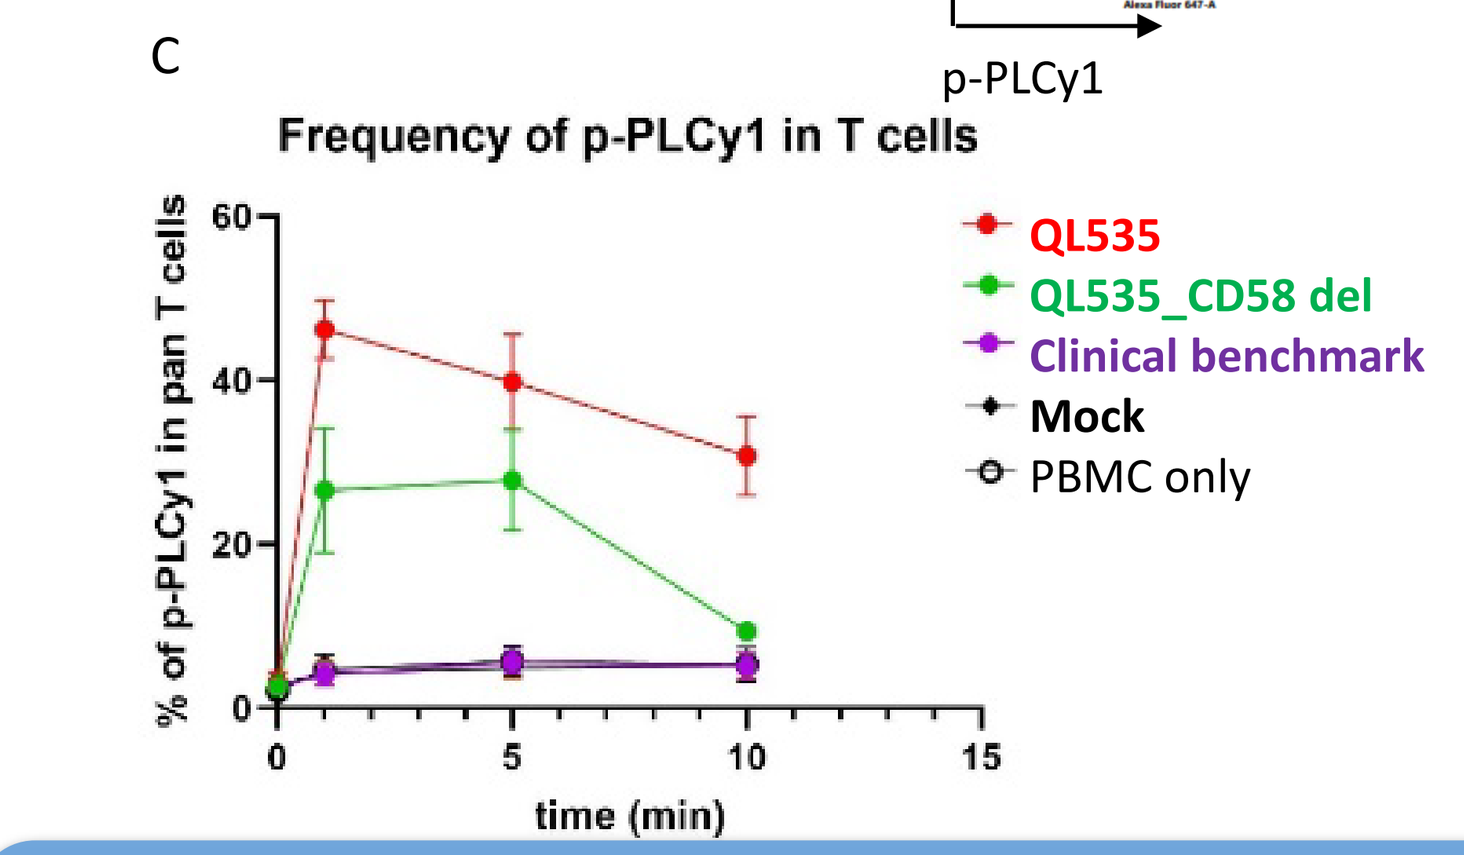
C
Frequency of p-PLCy1 in T cells
% of p-PLCy1 in pan T cells
time (min)
QL535
QL535_CD58 del
Clinical benchmark
Mock
PBMC only

Figure A. The experiment used barcode technology to differentiate samples from four time points to prevent batch effects. Figure B. Representative flow cytometry data comparing QL535 and the bispecific antibody. Figure C. Dot-line graphs of the percentage of pPLCγ1+ T cells for three individual donors and the combined data.

QL535 can Continuously Induce PBMCs in Prostate Cancer Patients Receiving Anti-PD1 Treatment to Sustain Cytotoxicity Against Tumor Cells *in vitro*.

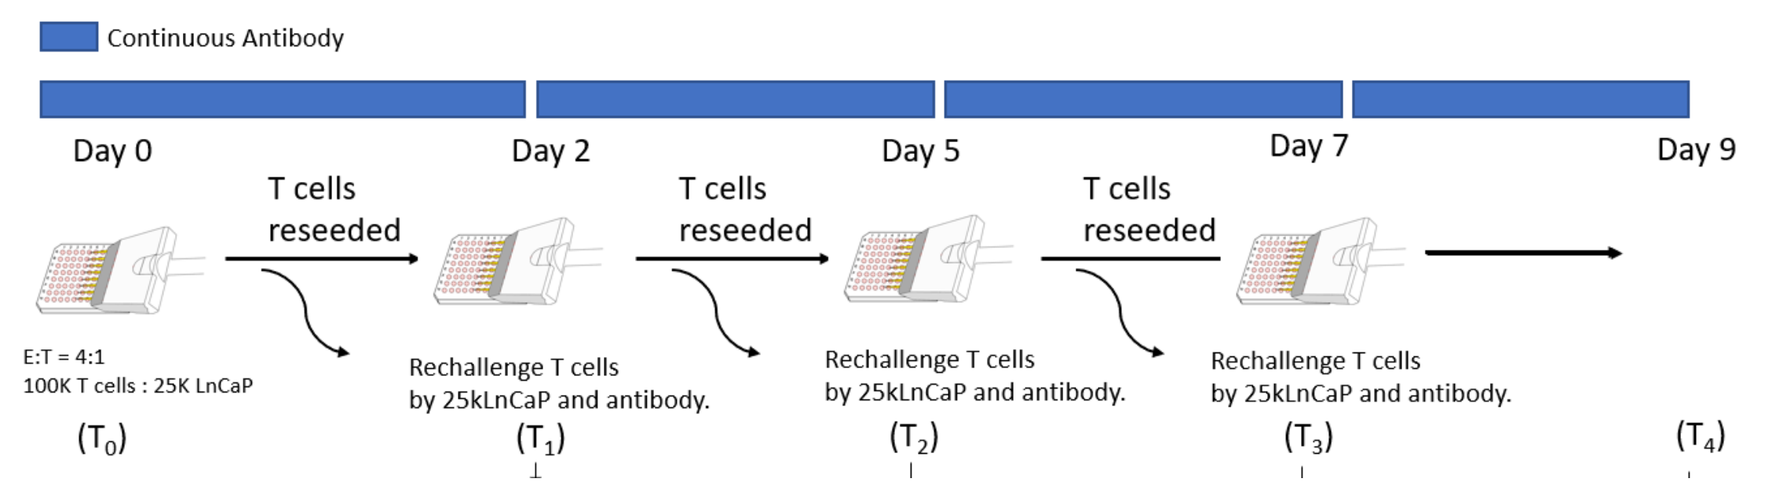
Continuous Antibody
Day 0
Day 2
Day 5
Day 7
Day 9
T cells reseeded
E:T = 4:1
100K T cells : 25K LnCaP
Rechallenge T cells by 25kLnCaP and antibody.
(T0)
(T1)
(T2)
(T3)
(T4)

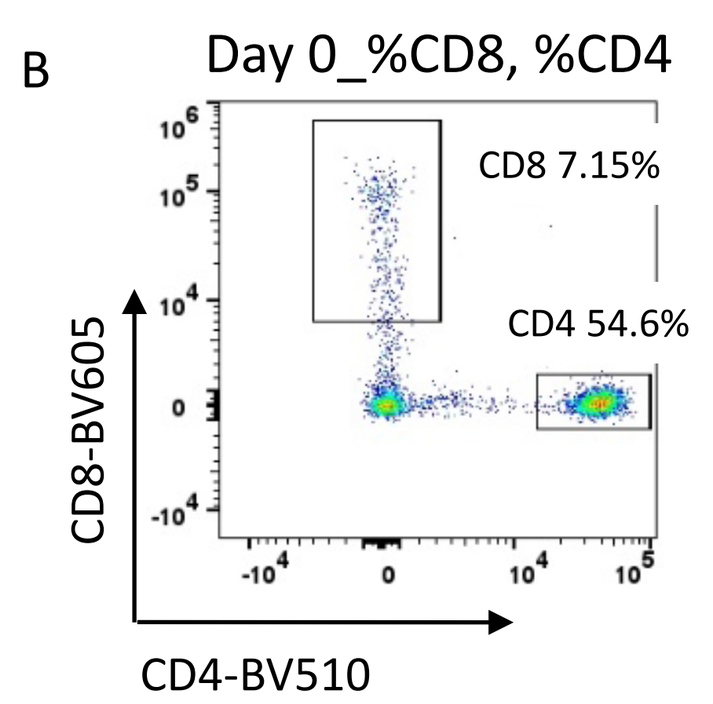
B
Day 0_%CD8, %CD4
CD8 7.15%
CD4 54.6%
CD8-BV605
CD4-BV510

A

Patient	Patient ID	Primary Diagnosis Name	Overall Clinical Stage	Treatment Status
Patient 1	122035464	Prostate Cancer, Adenocarcinoma	II	Opdivo

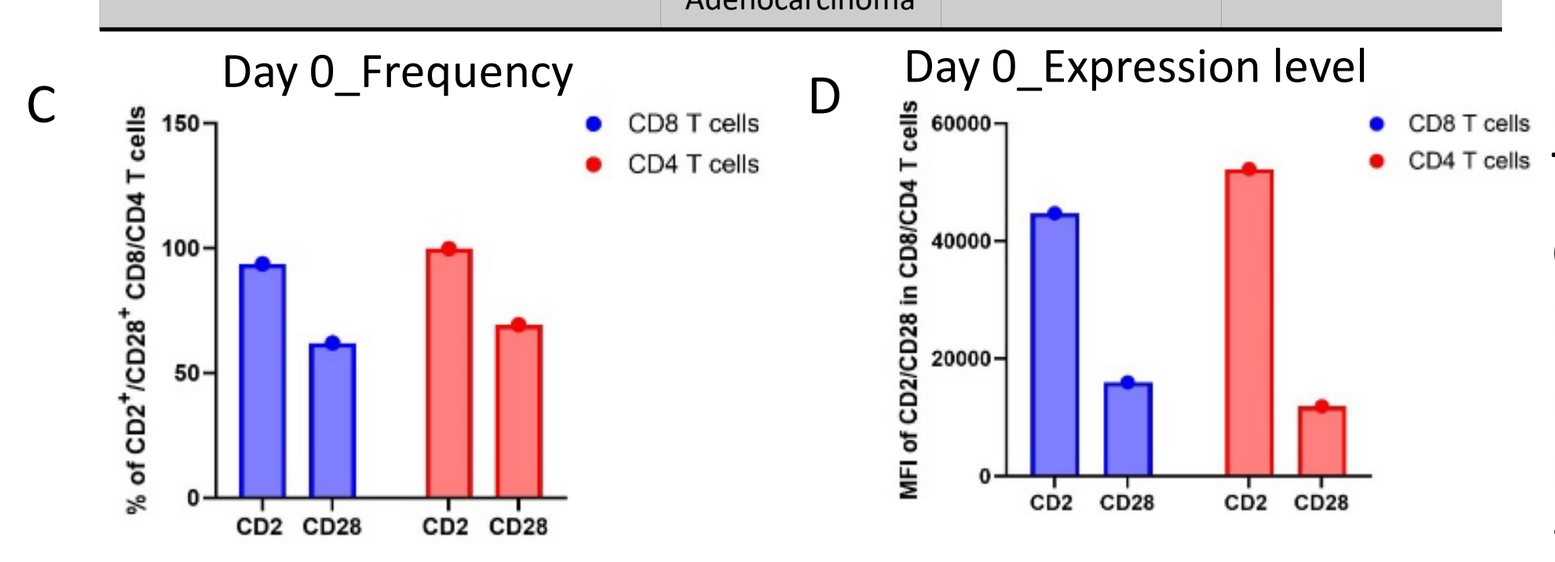
C
Day 0_Frequency
D
Day 0_Expression level
CD8 T cells
CD4 T cells
CD2
CD28

Table A. The clinical information of patient 1. Figure B. Patient 1's CD8 T-cell and CD4 T-cell percentages in total live PBMCs on day 0. Figure C. The frequency of CD2⁺/CD28⁺ CD4/CD8 T cells on day 0. Figure D. CD2/CD28 expression level (MFI) in CD4/CD8 T cells on day 0. Figure E. CellTilter readout of serial PBMC-based 22Rv1-specific killing *in vitro*. E:T=4:1. Time frames are day 4, day 7, day 10 and day 13.

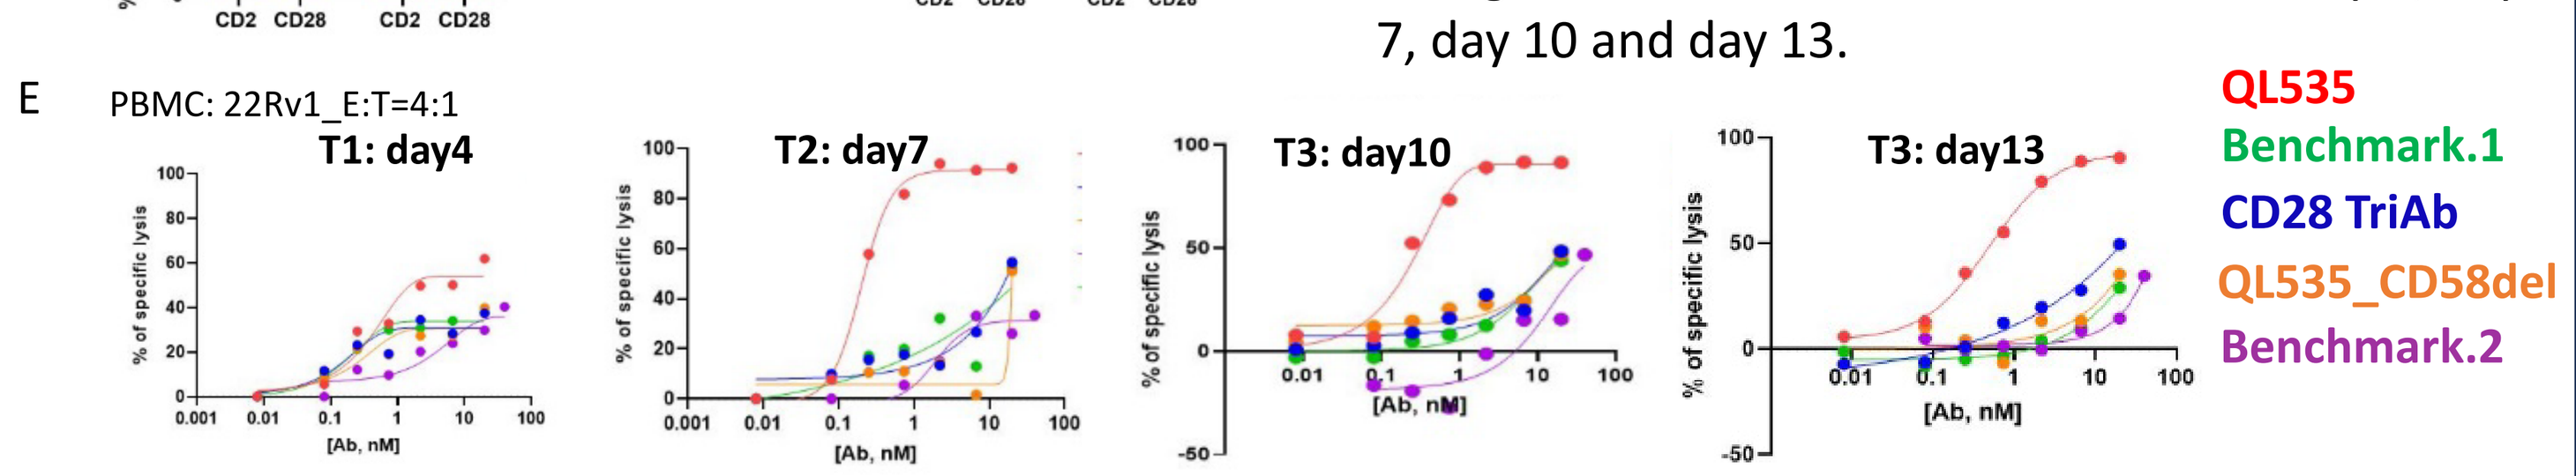
E
PBMC: 22Rv1_E:T=4:1
T1: day4
T2: day7
T3: day10
T3: day13
% of specific lysis
[Ab, nM]
QL535
Benchmark.1
CD28 TriAb
QL535_CD58del
Benchmark.2

No Significant Cytokine Release is Detected When PBMCs are Co-Incubated with QL535 *in vitro*

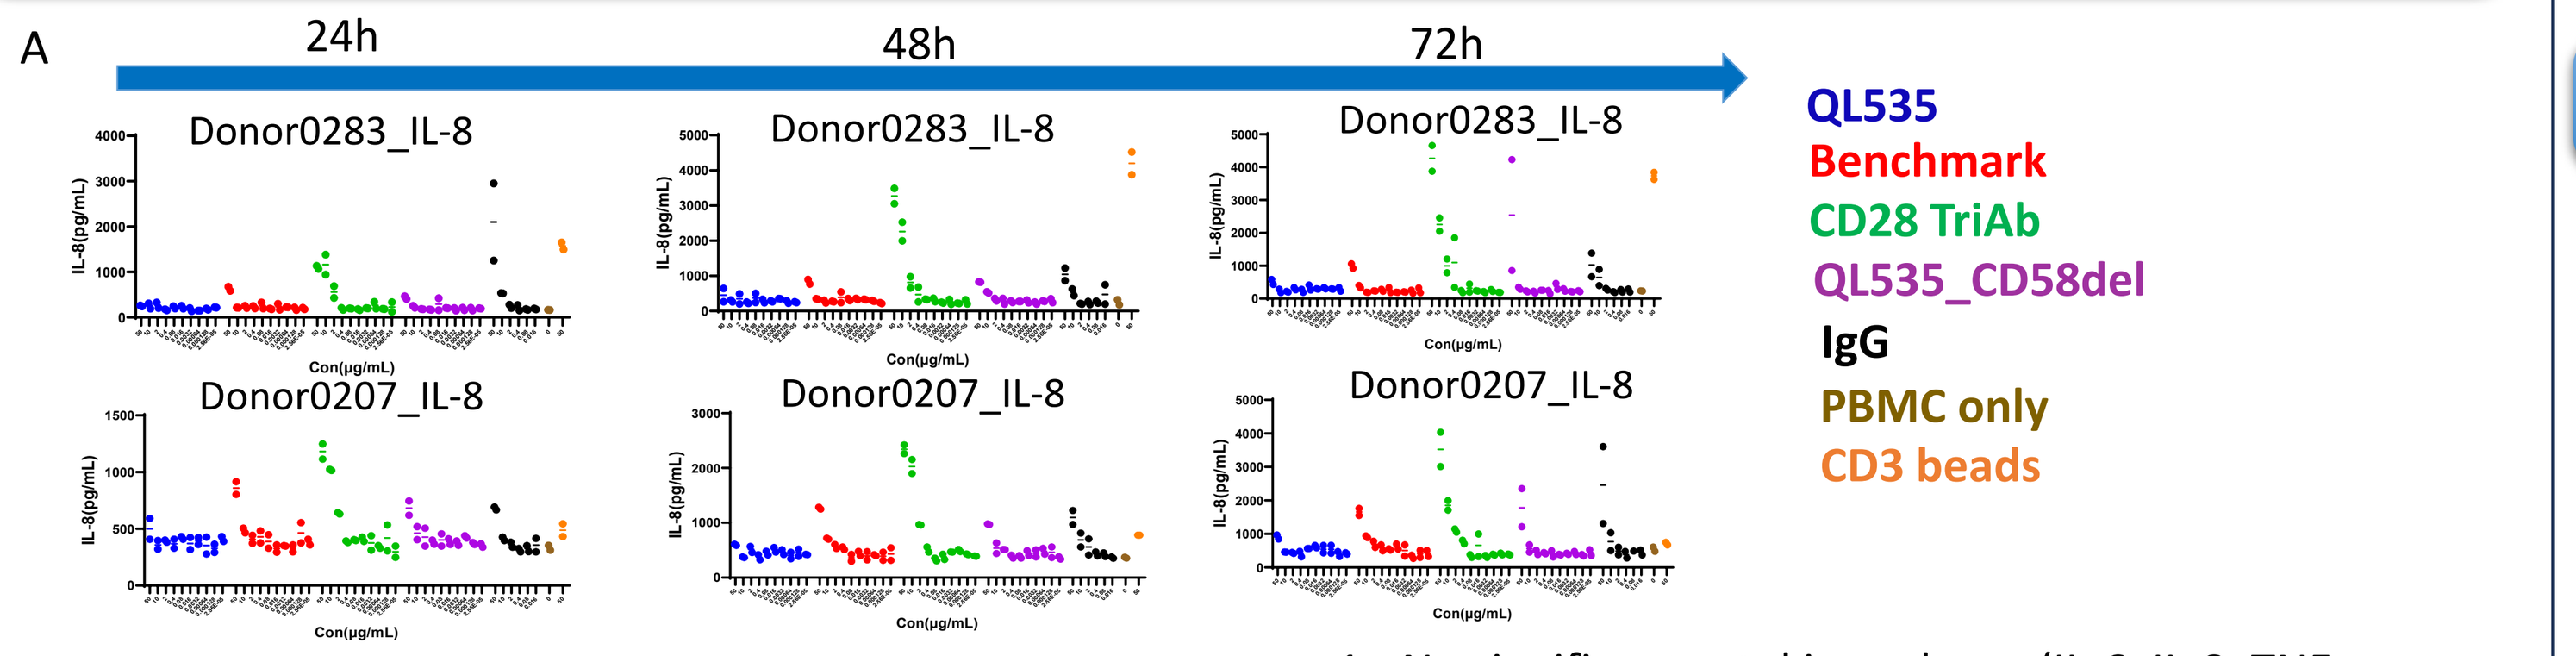
A
24h
48h
72h
Donor0283_IL-8
Donor0207_IL-8
Con(μg/mL)
QL535
Benchmark
CD28 TriAb
QL535_CD58del
IgG
PBMC only
CD3 beads

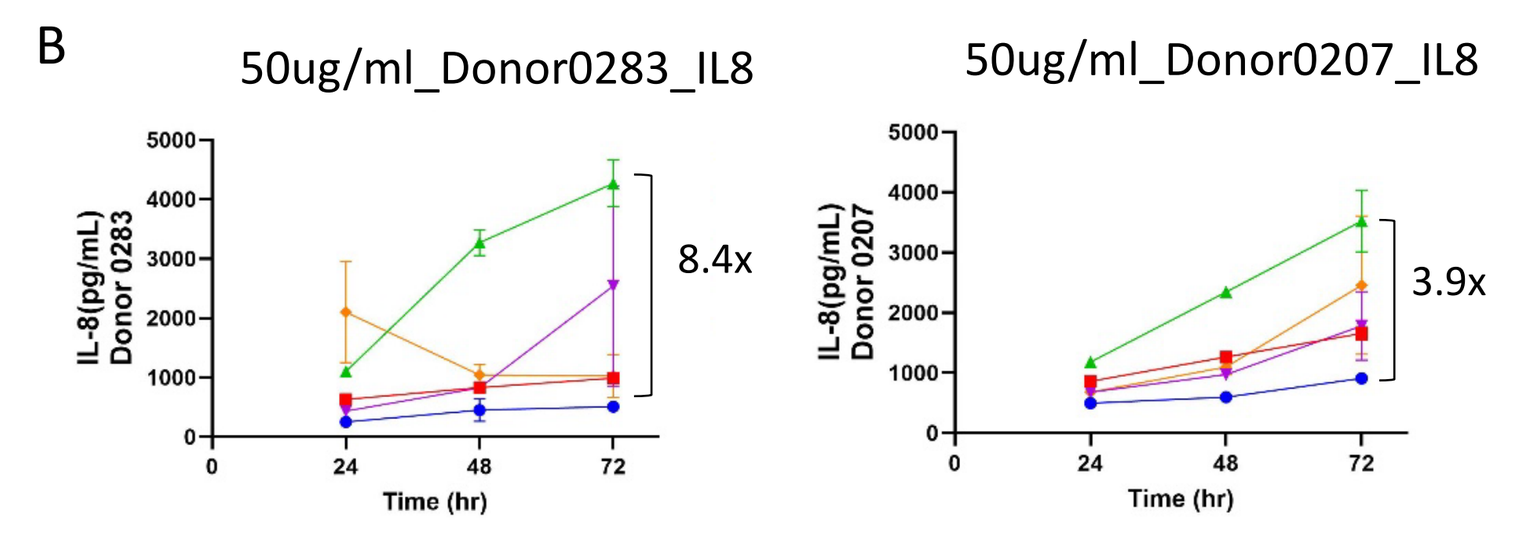
B
50ug/ml_Donor0283_IL8
50ug/ml_Donor0207_IL8
8.4x
3.9x
Time (hr)

1. No significant cytokine release (IL-6, IL-8, TNF**α**, IFN-**Y**, IL-10 and IL-2) was detected when PBMCs were co-Incubated with QL535 *in vitro.* We presented IL-8 data here.
2. However, both the CD28 trispecific antibody and the benchmark demonstrated cytokine release after incubation with PBMCs.

Superior Efficacy of QL535 in Xenografted NOG Mouse Model with 22RV1 Tumors

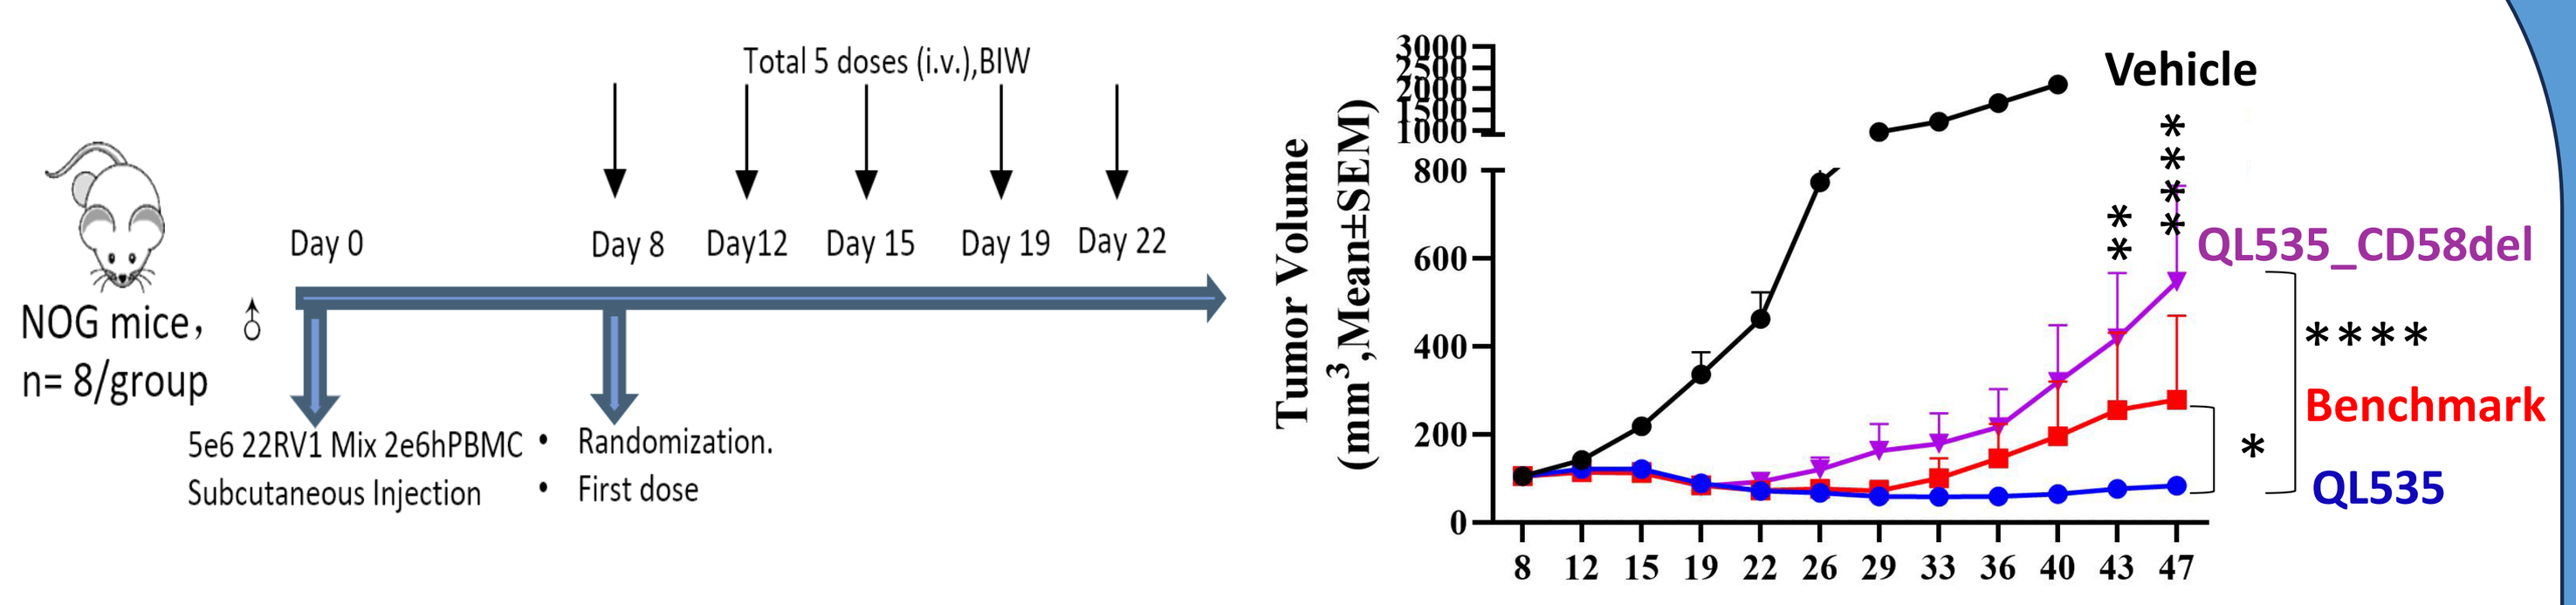
NOG mice, ♂
n= 8/group
Total 5 doses (i.v.),BIW
Day 0
Day 8
Day12
Day 15
Day 19
Day 22
5e6 22RV1 Mix 2e6hPBMC Subcutaneous Injection
Randomization.
First dose
Tumor Volume (mm³,Mean±SEM)
Vehicle
QL535_CD58del
Benchmark
QL535

*

In the *in vivo* efficacy comparison study in a NOG mouse xenograft model of 22RV1 tumors (with medium to low PSMA expression), QL535 demonstrated superior efficacy compared to the bispecific antibodies and clinical benchmark.

Superior Efficacy of QL535 in LNCAP-Luc NOG Mouse Orthotopic Transplant Model

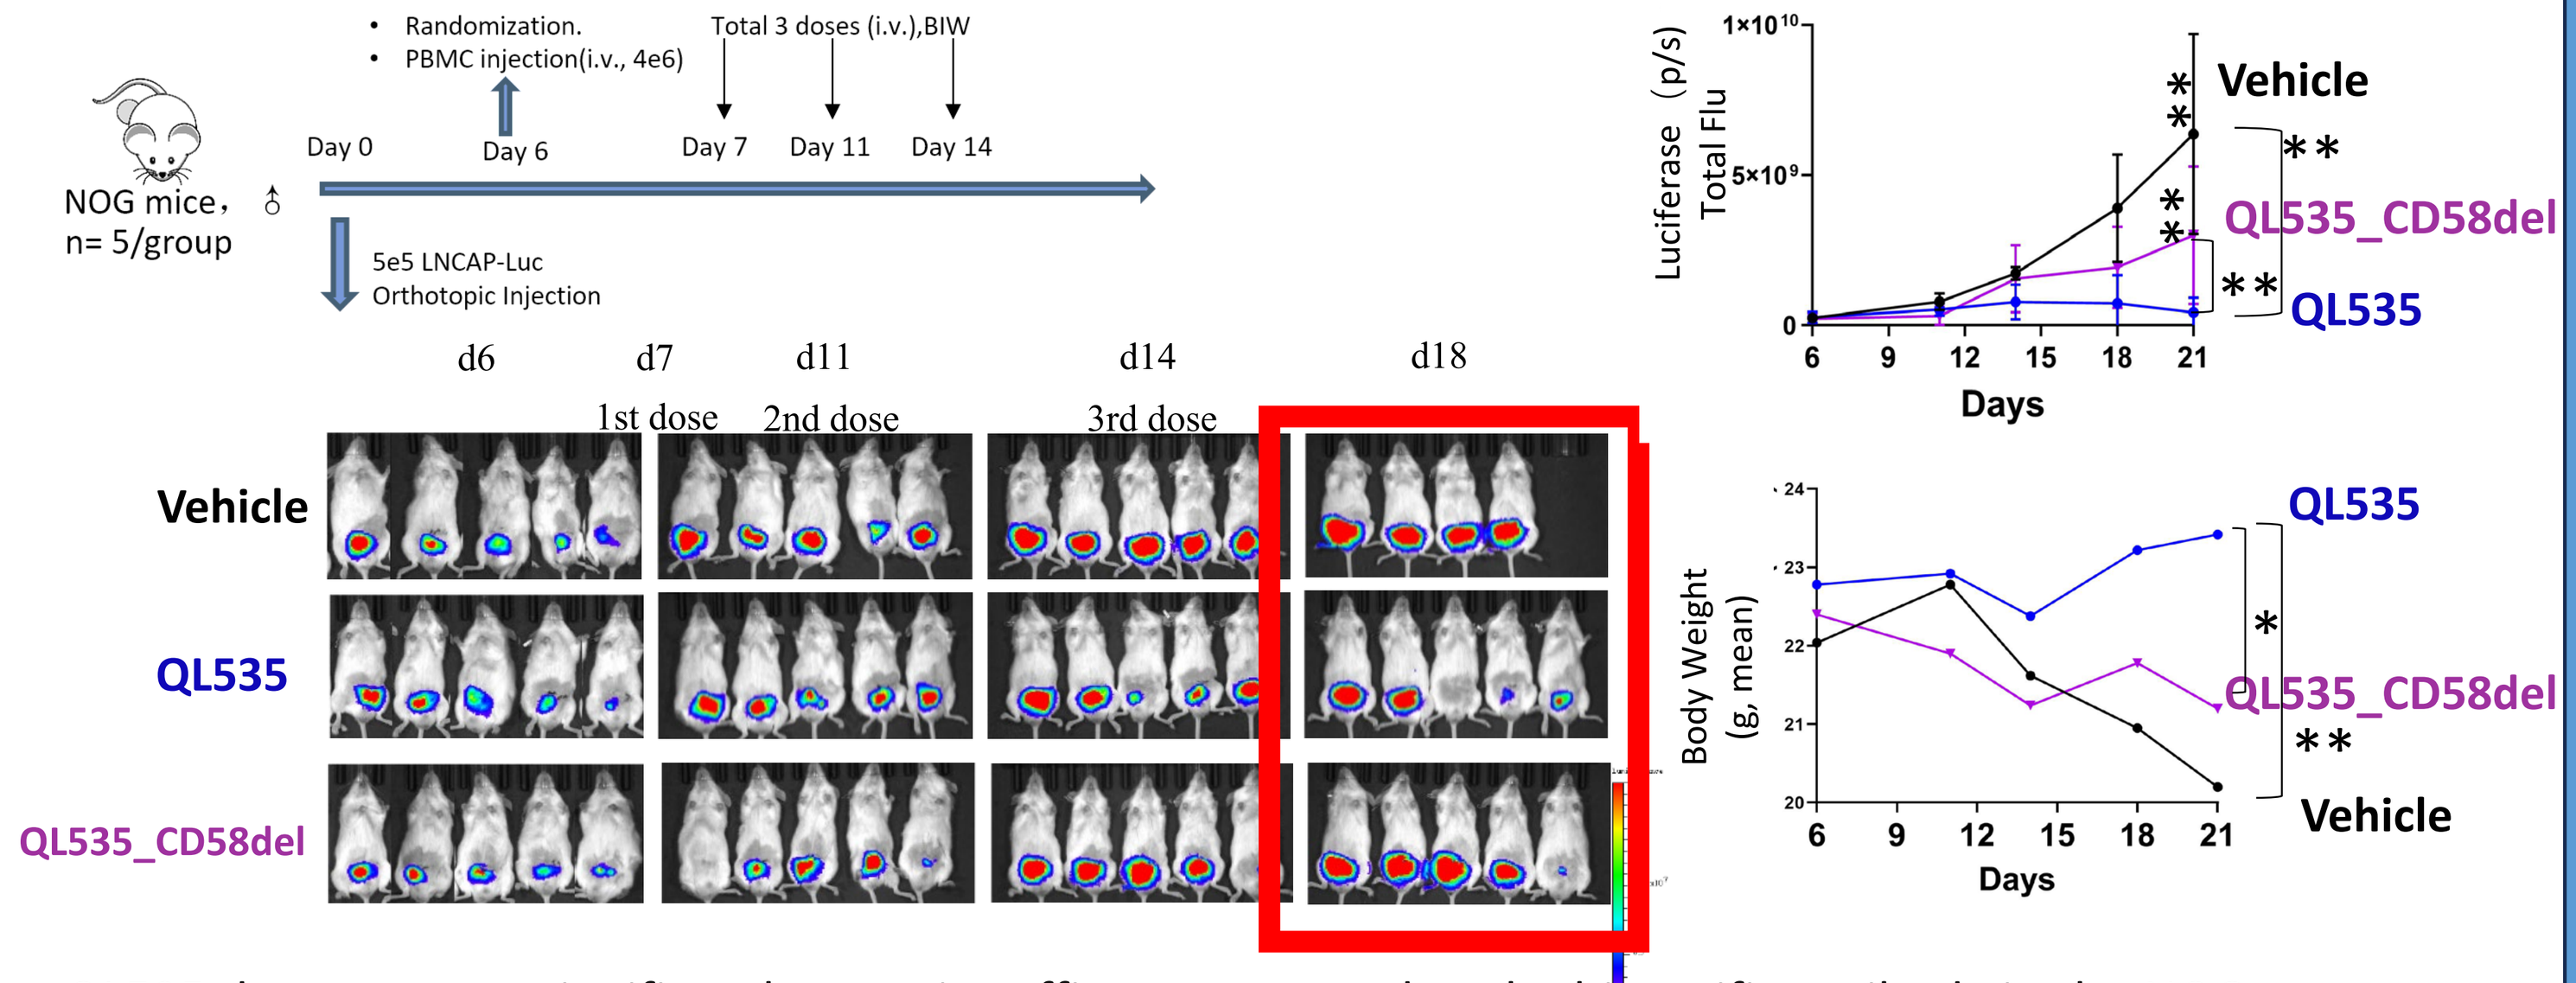
NOG mice, ♂
n= 5/group
Randomization.
PBMC injection(i.v., 4e6)
Total 3 doses (i.v.),BIW
Day 0
Day 6
Day 7
Day 11
Day 14
5e5 LNCAP-Luc Orthotopic Injection
d6
d7
d11
d14
d18
1st dose
2nd dose
3rd dose
Vehicle
QL535
QL535_CD58del
Luciferase (p/s) Total Flu
Body Weight (g, mean)
Days

QL535 demonstrates significantly superior efficacy compared to the bispecific antibody in the NOG mouse orthotopic transplant model of LNCAP-Luc (high PSMA expression) human prostate cancer.

Activation of T Cells in PBMCs by Serum from Cynomolgus Monkeys After Single Dosing of QL535

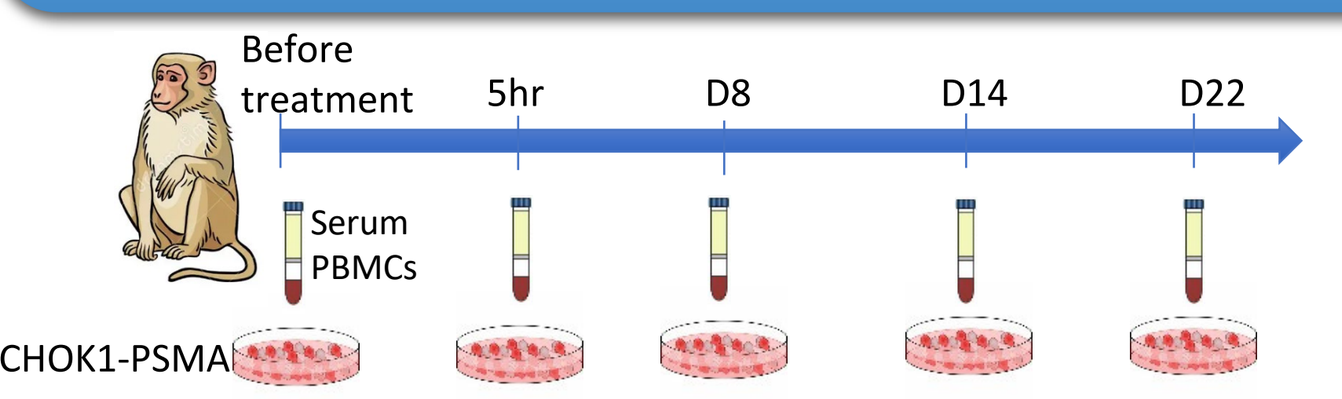
Before treatment
5hr
D8
D14
D22
Serum PBMCs
CHOK1-PSMA

❑ At each time point, collect cynomolgus monkey PBMCs + serum + CHOK1-PSMA cell line to co-incubate, measuring T-cell activation; before medication, 5 hours, Day 8, Day 14, Day 22 after medication.

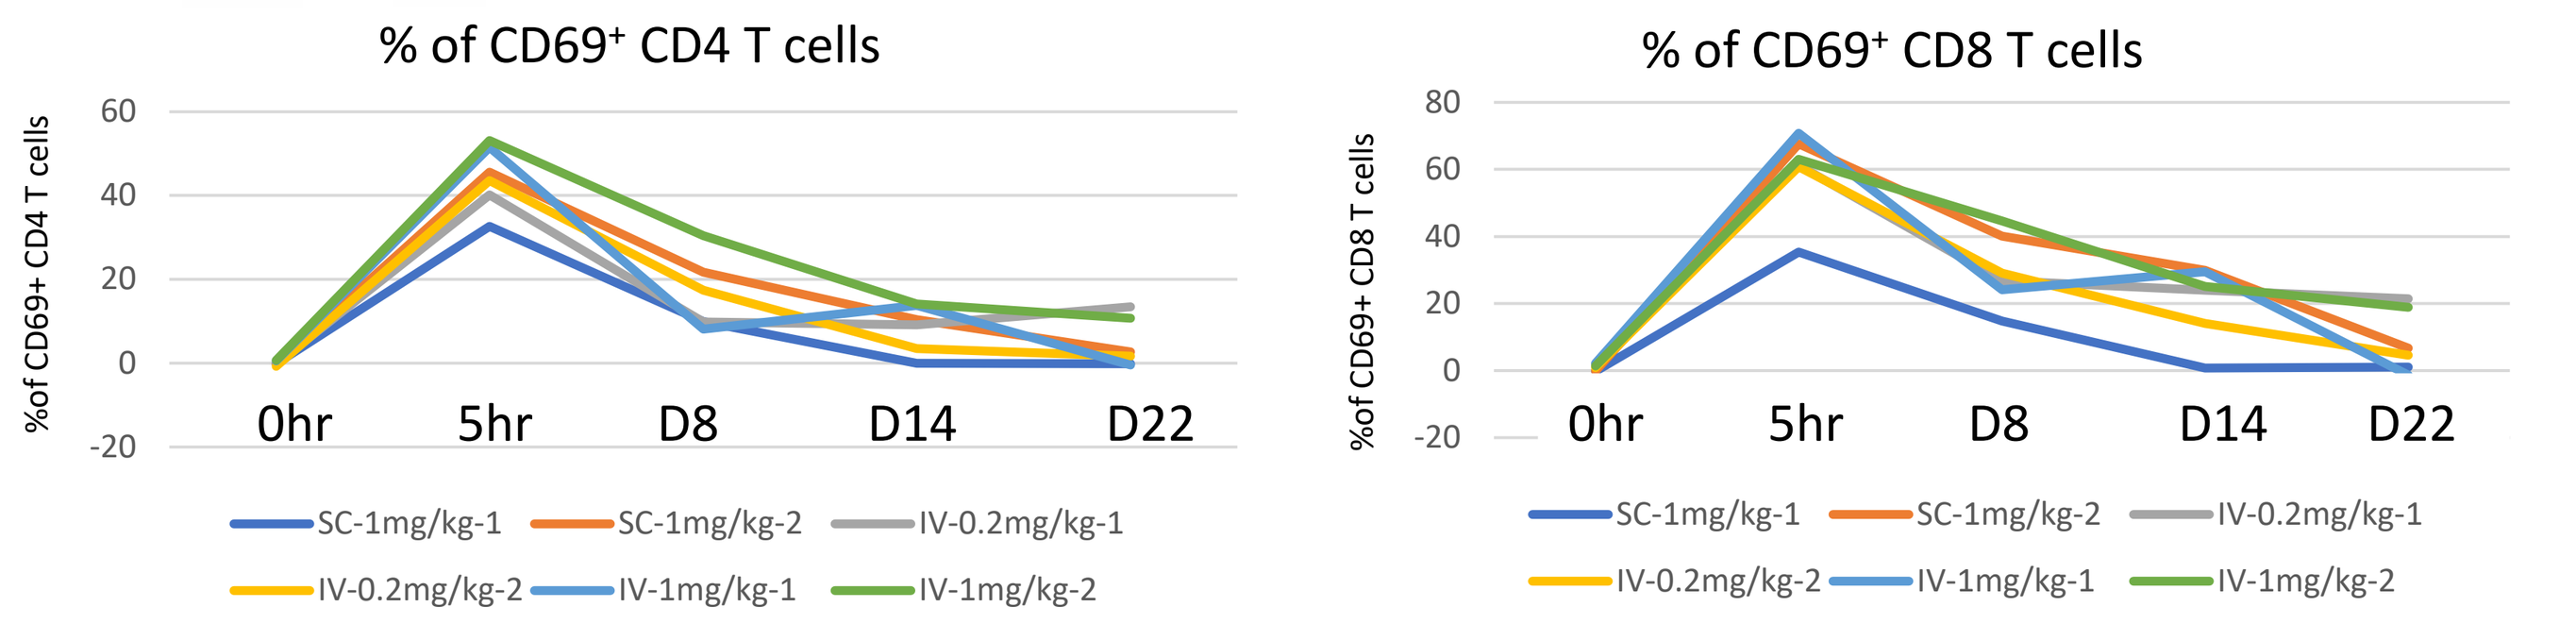
% of CD69⁺ CD4 T cells
% of CD69⁺ CD8 T cells
0hr
5hr
D8
D14
D22
SC-1mg/kg-1
SC-1mg/kg-2
IV-0.2mg/kg-1
IV-0.2mg/kg-2
IV-1mg/kg-1
IV-1mg/kg-2

Regardless of whether administered via IV or SC, after a single dose of QL535 in cynomolgus monkeys, T cells in PBMCs can be activated by serum, which were extracted at 5hr time point, with co-cultured PSMA-positive cell lines *in vitro.*

Conclusions

- **Target Specificity and Costimulation**: QL535, a TECOS with CD2 costimulation, specifically targets PSMA-positive prostate cancer, offering an advantageous approach where CD28 costimulatory factors are minimally expressed at the tumor site.
- **Therapeutic Developability**: QL535 demonstrates favorable therapeutic developability, indicating its potential for efficient and scalable production and clinical application.
- **Efficacy and Safety Profile**: QL535 achieves the same *in vitro* killing efficacy as the CD28 trispecific antibody, but with an improved safety profile characterized by reduced cytokine release.
- **Mechanism of Action**: The mechanism of action of QL535 involves the amplification of the pPLC-γ1 signaling pathway, enhancing its therapeutic effect.
- **Persistent Activation**: QL535 is capable of repeatedly stimulating prostate cancer patient PBMCs, enabling them to continuously target and eliminate PSMA+ cancer cells.
- ***In Vivo* Efficacy**: Demonstrated excellent efficacy in *in vivo* models, further validating QL535's potential as an effective treatment for prostate cancer.
- **Immune Activation**: Serum from cynomolgus monkeys following a single dose of QL535 can activate T-cells antigen-dependently *in vitro*, underscoring its potential to enhance immune responsiveness against tumors.

Disclosures

X Liu, S Gu, J Morrissette, G Guo, J Chen, N Ngo, S Chen are current or former employees of QLSF Biotherapeutics, Inc. receiving salaried compensation and stock options.
K Liu, J Kong, Y Wang, X Zhang, J Cao, M Hu, M Sun, R Ma, H Zhu, L Wang, Y Fang, H Liu, S Zhao are current employees of Qilu Pharmaceutical Co., Ltd. receiving salaried compensation.

Selected References

1. Sung, H., Ferlay, J., Siegel, R. L., Laversanne, M., Soerjomataram, I., Jemal, A., & Bray, F. (2021). Global cancer statistics 2020: GLOBOCAN estimates of incidence and mortality worldwide for 36 cancers in 185 countries. *CA: A Cancer Journal for Clinicians*, 71(3), 209–249.
2. American Cancer Society. (2025). *Key statistics for prostate cancer*.
3. Scherer, K., Kumar, A., & Lütje, S. (2020). "Prostate-Specific Membrane Antigen: Advancements in Targeted Therapy and Imaging Modalities." *Journal of Nuclear Medicine*, 61(1), 82-89.